FINDING SUCCESS WITH ACTIVE *LEPTOSPERMUM* HONEY AFTER FAILURES WITH NEGATIVE PRESSURE WOUND THERAPY

Aaron Wodash RN, WCC
Augustana Health Care Center, Minneapolis, Minnesota

INTRODUCTION

The goal of achieving wound closure is often challenging with complicated patients suffering with multiple co-morbidities. Even when choosing advanced wound care dressings and therapies, often times thousands of dollars are spent without progress. This series describes patients suffering with wounds that were stalled in their progression toward healing despite receiving negative pressure wound therapy (NPWT). NPWT was initially chosen because of the large size wounds and significant amounts of exudate. However, they were still not showing improvement when an alternative therapy was decided upon.

METHODS

A change in the plan of care was necessary and it was decided to try Active *Leptospermum* Honey (ALH)* dressings for their known mechanism to jump-start the wound healing process. ALH dressings are known to have a low pH important to alter the environment of the wound bed, and believed to be one of the mechanisms responsible for faster wound healing.[1] ALH has also been shown to promote autolytic debridement,[2] and in our facility has often been used in lieu of enzymatic debriding agents.

The patients in this series had many challenging issues and medical complications including malnourishment, contractures, and the inability to communicate. Patient #1 presented with three stage IV pressure ulcers being treated with NPWT in combination with an enzymatic debriding agent 3x/week for a period of three months but was not showing expected progress. ALH Gel was first initiated in combination with NPWT and the remaining area of slough in the wound was quickly debrided after one week and the wound then began to reduce in size. The NPWT was able to be discontinued and ALH was continued in combination with a Super Absorbent dressing** being changed twice weekly. The rapid progress after months of little progress was considered remarkable.

Patient #2 presented with a full thickness surgical wound also not demonstrating improvement with NPWT. It was decided to discontinue the NPWT and trial ALH after seeing the rapid response in patient #1. This wound also rapidly began to reduce in size and showed significant improvement with the use of ALH.

PATIENT 1

A 79 yo female patient was living at home with family receiving home health nursing services and she developed multiple stage IV pressure ulcers. Home health service stopped seeing patient because she required more care than they could provide. Pt was hospitalized for wound infection, UTI, bacteremia, dehydration, and severe protein-calorie malnutrition. Patient underwent incision and drainage with NPWT placement at the hospital and was admitted to our facility for LTC. Past medical history includes CVA x2, DM II, anemia, hyperlipidemia, HTN, and severe bilateral hip and knee contractures. Patient was bedfast, non-verbal, and immobile.

On 2/29/12, patient presented with stage IV Left Ischial tuberosity (Left IT), Right ischial tuberosity (Right IT) and Left Trochanter ulcers, which were stalled. The Left IT ulcer had been treated previously with collaginase and NPWT 3x per week x 3 months. For a short period of time ALH Gel was trialed under NPWT and after improvement NPWT was discontinued and the plan was changed to ALH Calcium Alginate with Super Absorbent dressing 2x per week. A slough plug covering 5% of wound bed debrided after one week of ALH treatments being used. ALH started 3/7/12 and was utilized until complete wound closure. The Right IT and Left Trochanter wounds also began to show marked improvement with the change in the plan of care to ALH.

Left IT

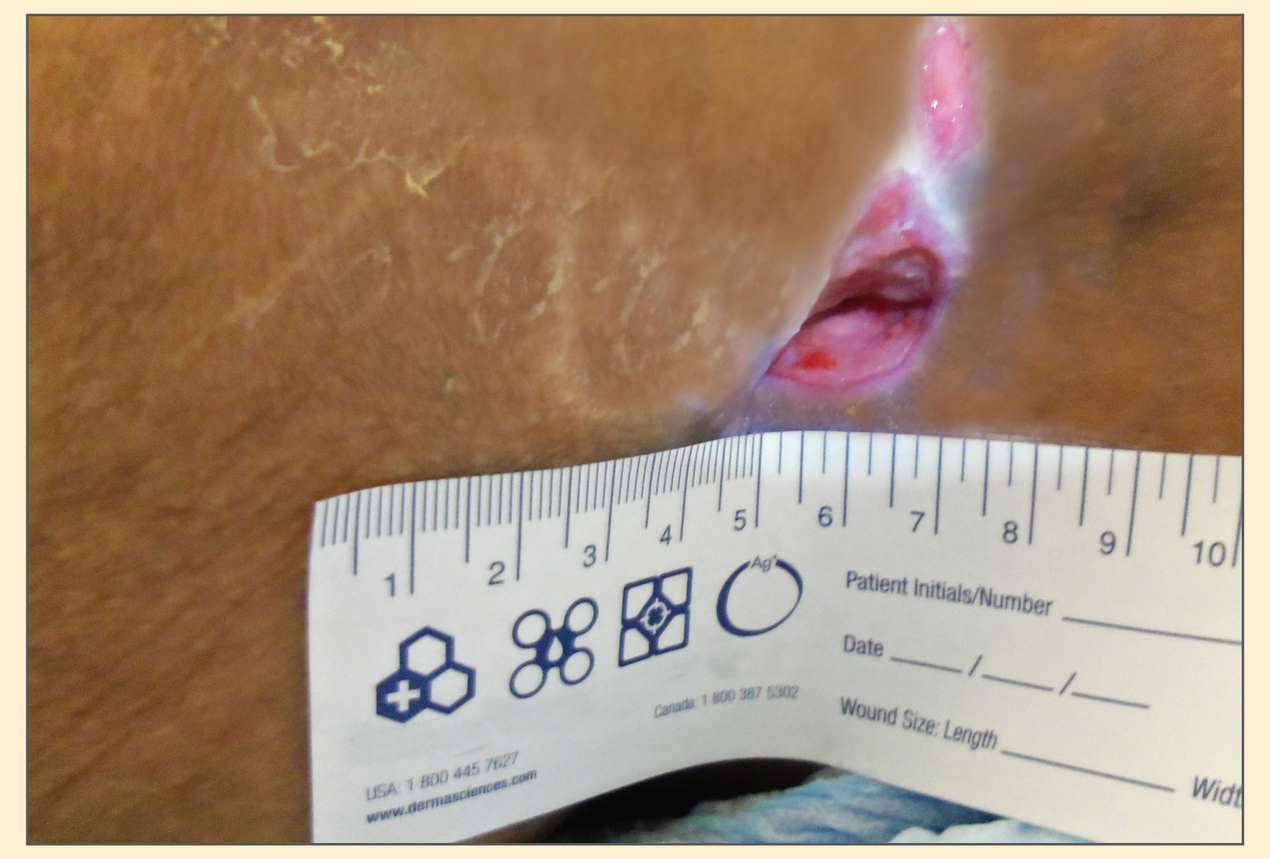
2/14/12

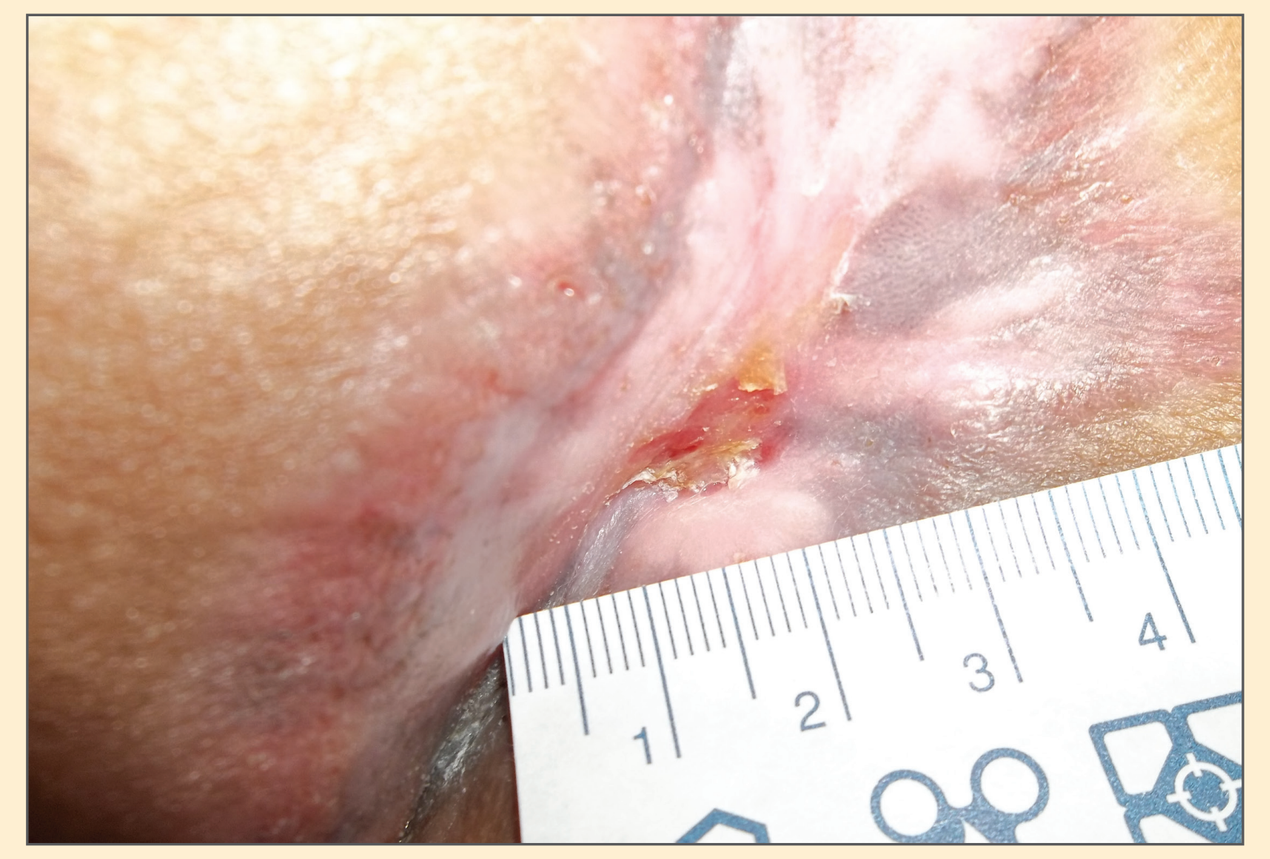
5/23/12

Date	Length	Width	Depth	Area (cm^2)	Volume (cm^3)	% Area Reduction	% Volume Reduction
2/29/12	4.0	2.0	4.0	6.28	25.13	Initial Measurement	Initial Measurement
3/7/12	2.9	2.5	3.2	5.69	18.22	9.4%	27.5%
3/14/12	2.7	1.8	3.2	3.82	12.21	39.3%	51.4%
3/21/12	2.6	1.9	2.1	3.88	8.15	38.3%	67.6%
3/28/12	1.8	1.3	1.8	1.84	3.31	70.8%	86.8%
4/3/12	1.5	2.0	1.8	2.36	4.24	62.5%	83.1%
4/11/12	1.2	1.3	1.0	1.23	1.23	80.5%	95.1%

Right IT

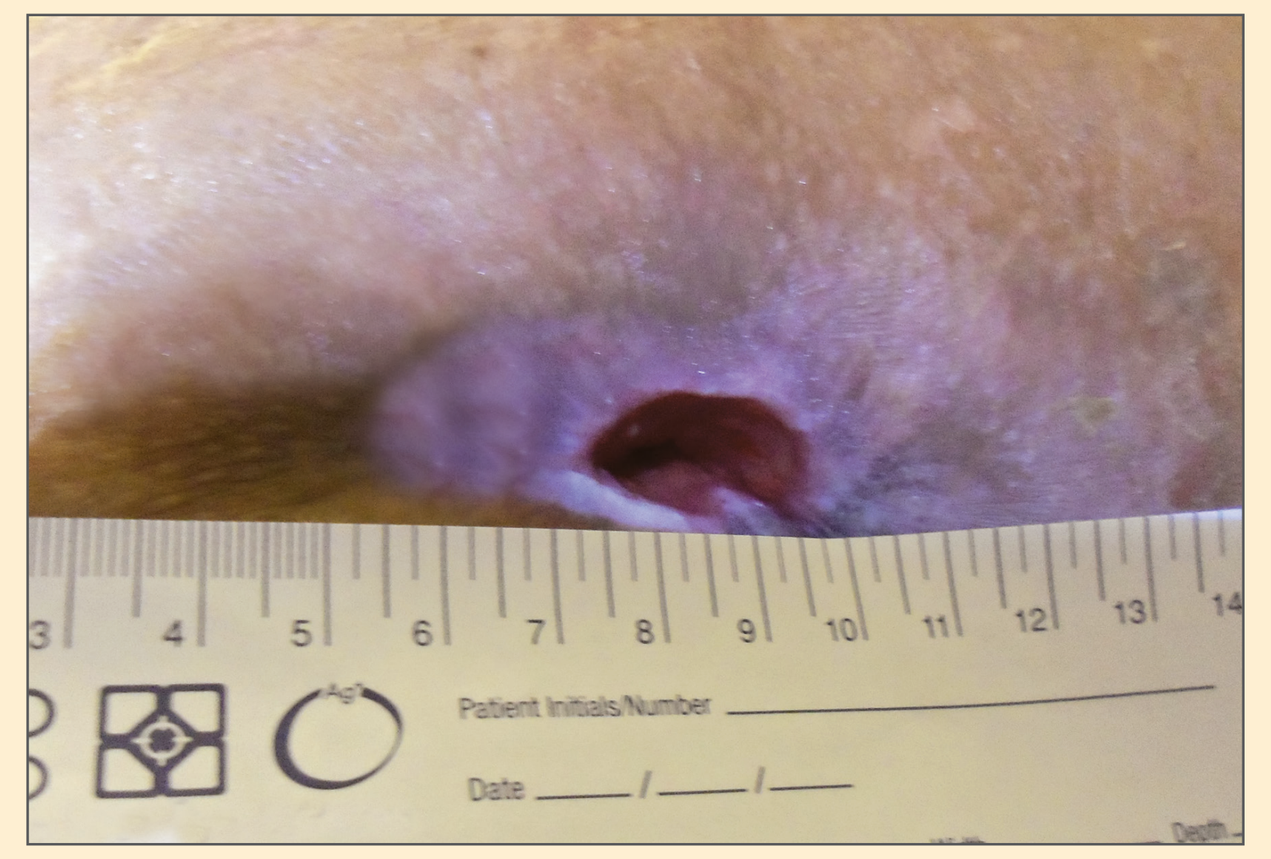
2/14/12

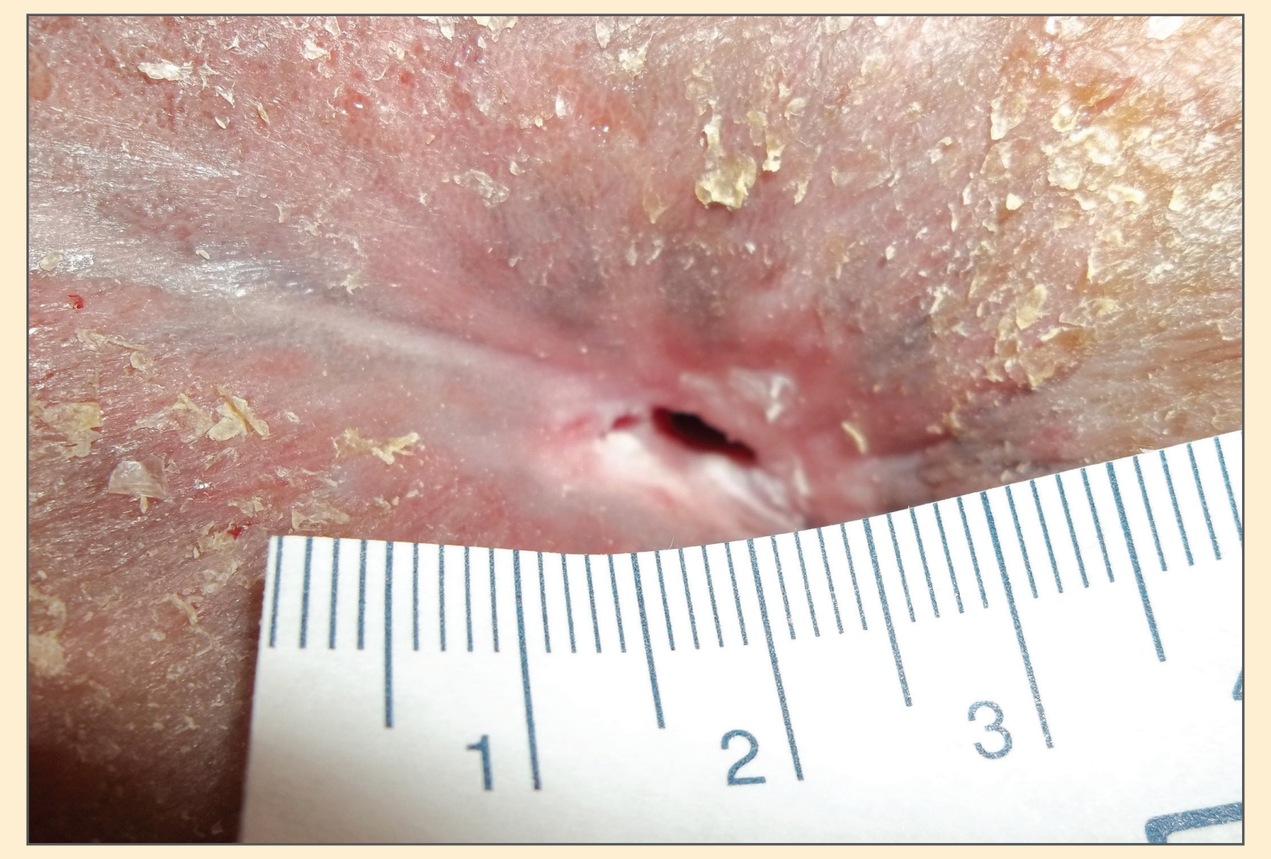
5/23/12

Date	Length	Width	Depth	Area (cm^2)	Volume (cm^3)	% Area Reduction	% Volume Reduction
2/29/12	1.7	2.2	5.3	2.94	15.57	Initial Measurement	Initial Measurement
3/7/12	1.8	1.7	4.4	2.40	10.57	18.2%	32.1%
3/14/12	1.7	1.9	4.3	2.54	10.91	13.6%	29.9%
3/21/12	1.6	2.1	4.0	2.64	10.56	10.2%	32.2%
3/28/12	1.7	1.7	3.5	2.27	7.94	22.7%	49.0%
4/3/12	1.4	1.8	3.5	1.98	6.93	32.6%	55.5%
4/11/12	1.2	1.4	2.8	1.32	3.69	55.1%	76.3%

Left Trochanter

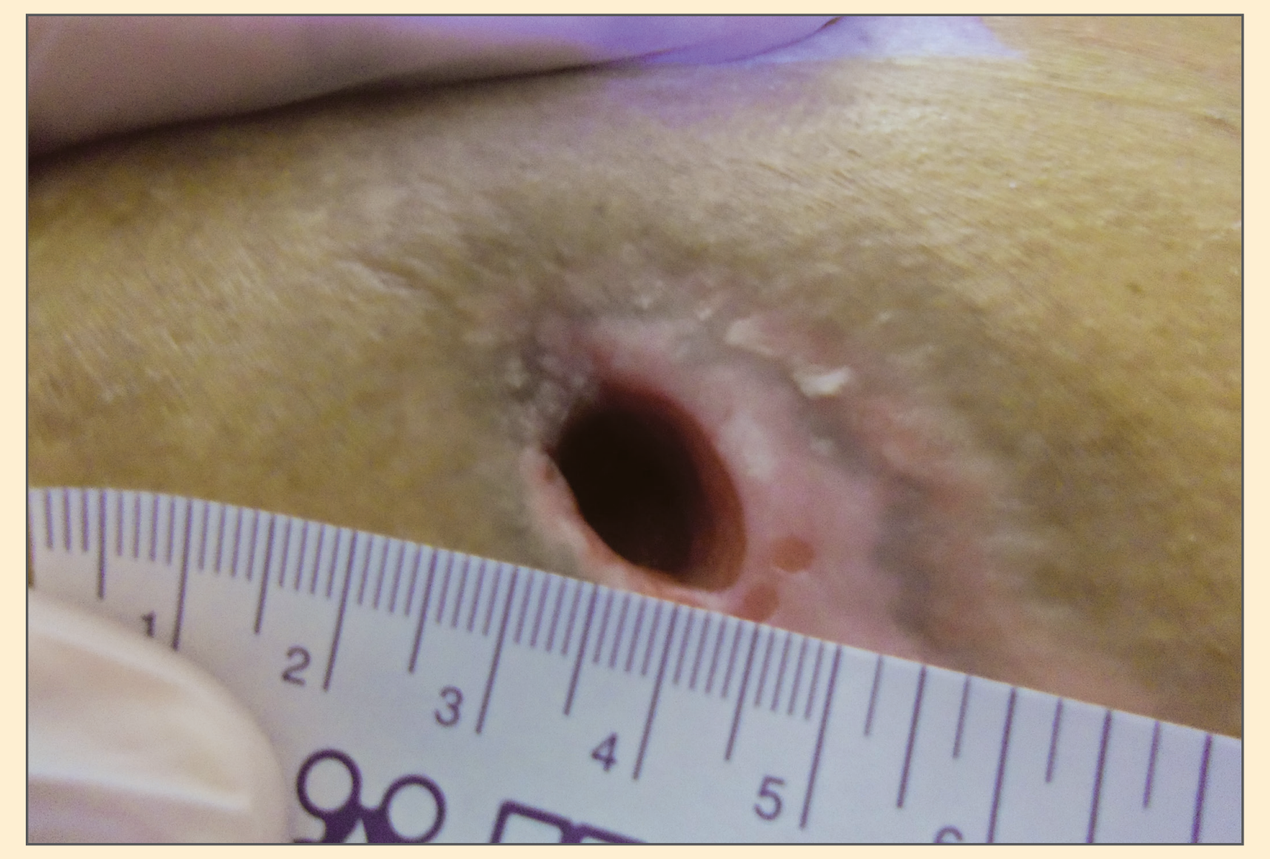
2/14/12

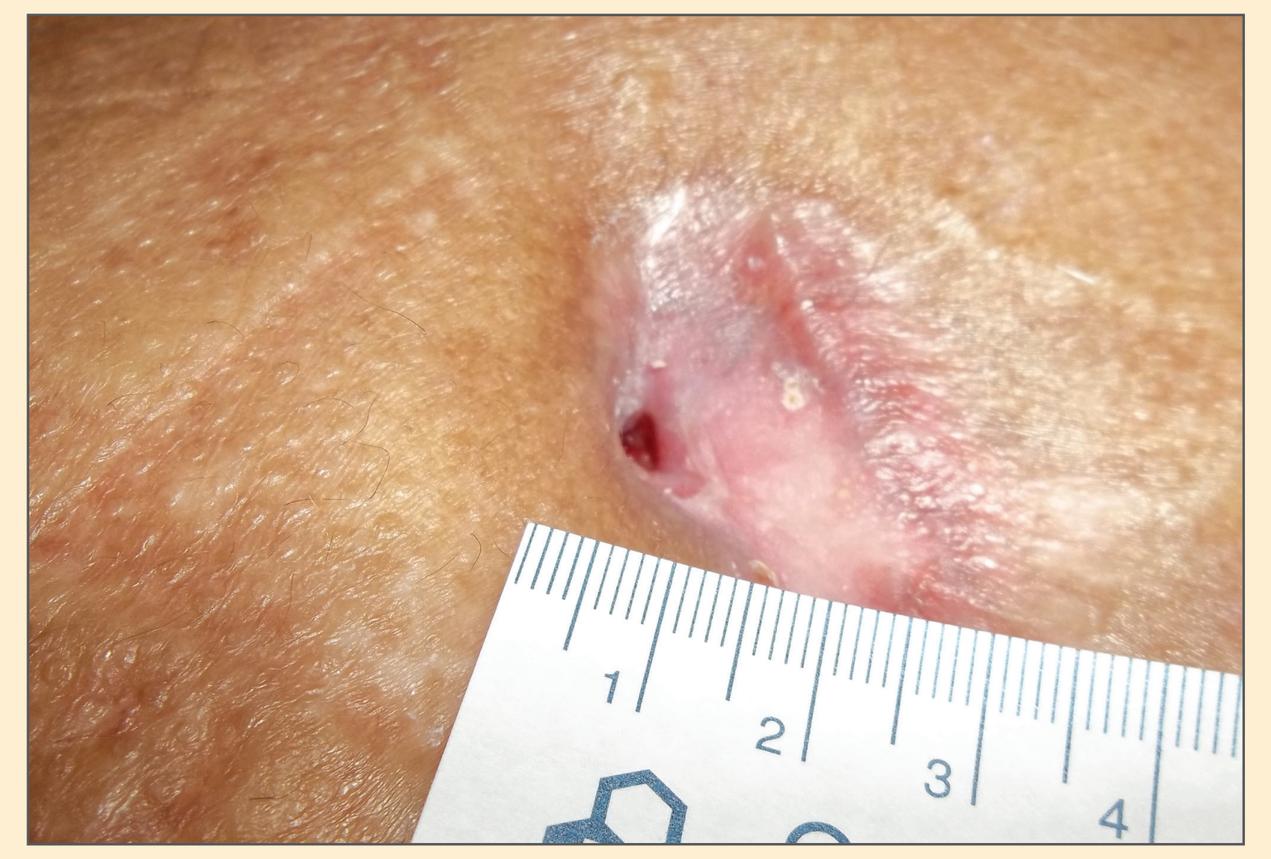
5/23/12

Date	Length	Width	Depth	Area (cm2)	Volume (cm3)	% Area Reduction	% Volume Reduction	Undermining	% UM Reduction
2/29/12	1.3	5.8	1.4	5.92	8.29	Initial Measurement	Initial Measurement	4.5	Initial Measurement
3/7/2012	1.3	4.5	0.8	4.59	3.68	22.4%	55.7%	3.2	28.9%
3/14/12	1.3	4.1	0.8	4.19	3.35	29.3%	59.6%	2.8	37.8%
3/21/12	1.0	3.6	0.6	2.83	1.70	52.3%	79.5%	2.7	40.0%
3/28/12	0.7	0.8	0.6	0.44	0.26	92.6%	96.8%	4.4	2.2%
4/3/12	0.7	0.7	0.1	0.38	0.04	71.0%	97.9%	3.5	22.2%
4/11/12	0.8	0.5	0.1	0.31	0.03	76.3%	98.3%	3.2	28.9%

Please note that the increase in undermining measurements were due to a change in measurement technique and documenting a more lateral depth/tunnel. There was not an increase in true depth but in fact was an attempt to more accurately capture the wound volume by measuring the tunnel at 10:00 to the deepest point.

PATIENT 2

Patient was an 81 year old female with multiple medical problems including: CVA, HTN, CAD, DM, hyperlipidemia, and fall with displaced R femoral neck fracture, s/p R hip hemiarthroplasty on 9/15/11. Pt was hospitalized for post surgical infection requiring explantation on 10/10/11 and then underwent incision and drainage with NPWT placement on 10/27/11. NPWT continued until 2/29/12 when the medical team changed the plan of care to a plain calcium alginate dressing and foam cover, due to the wound being stalled. Little progress was seen with the plain calcium alginate. On 3/4/12 ALH Calcium Alginate dressing was started with a Super Absorbent dressing as a secondary dressing and the wound again began to progress. On 3/19/12, an NP changed the plan of care to wet to dry dressings daily. Again the wound failed to progress and ALH was reinstated on 4/7/12 via an order from ortho. The wound continued to improve with this plan of care.

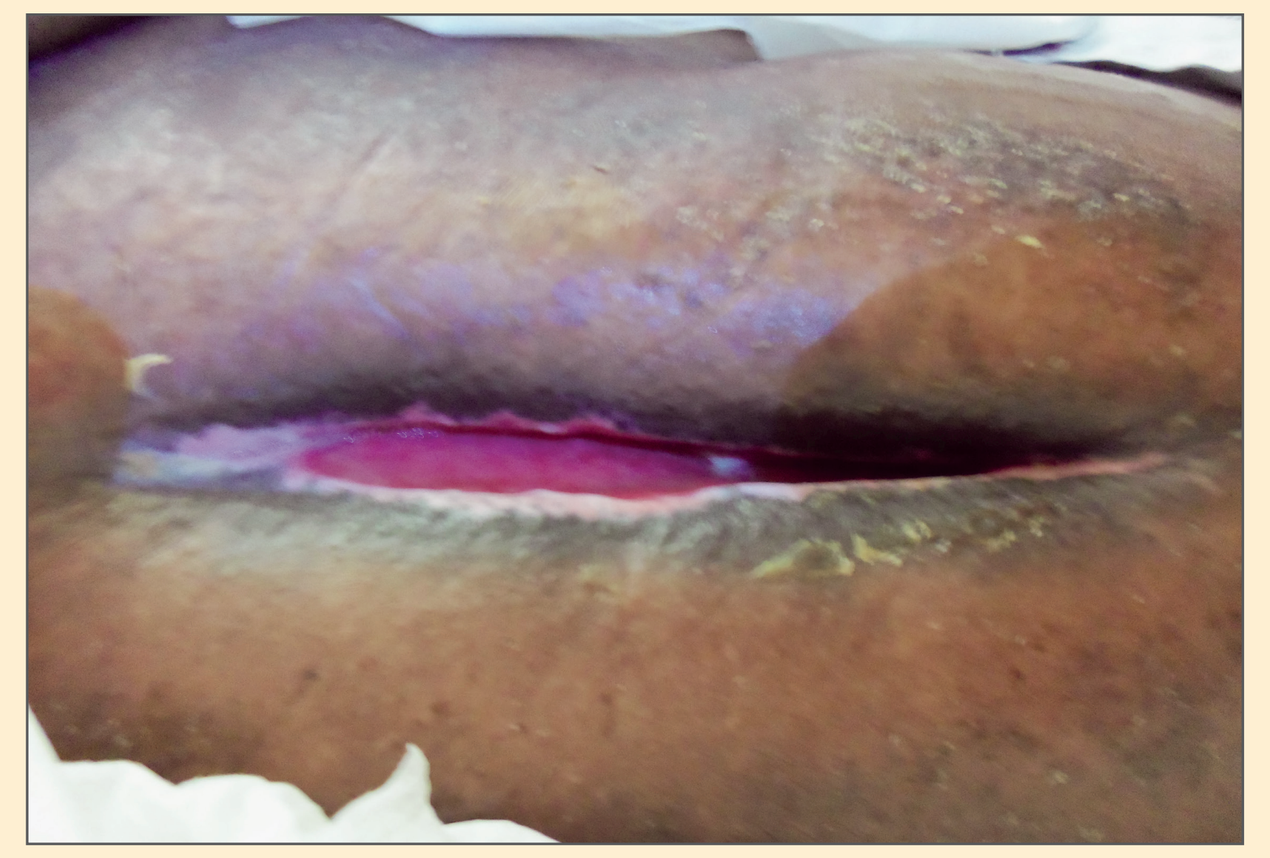
2/14/12

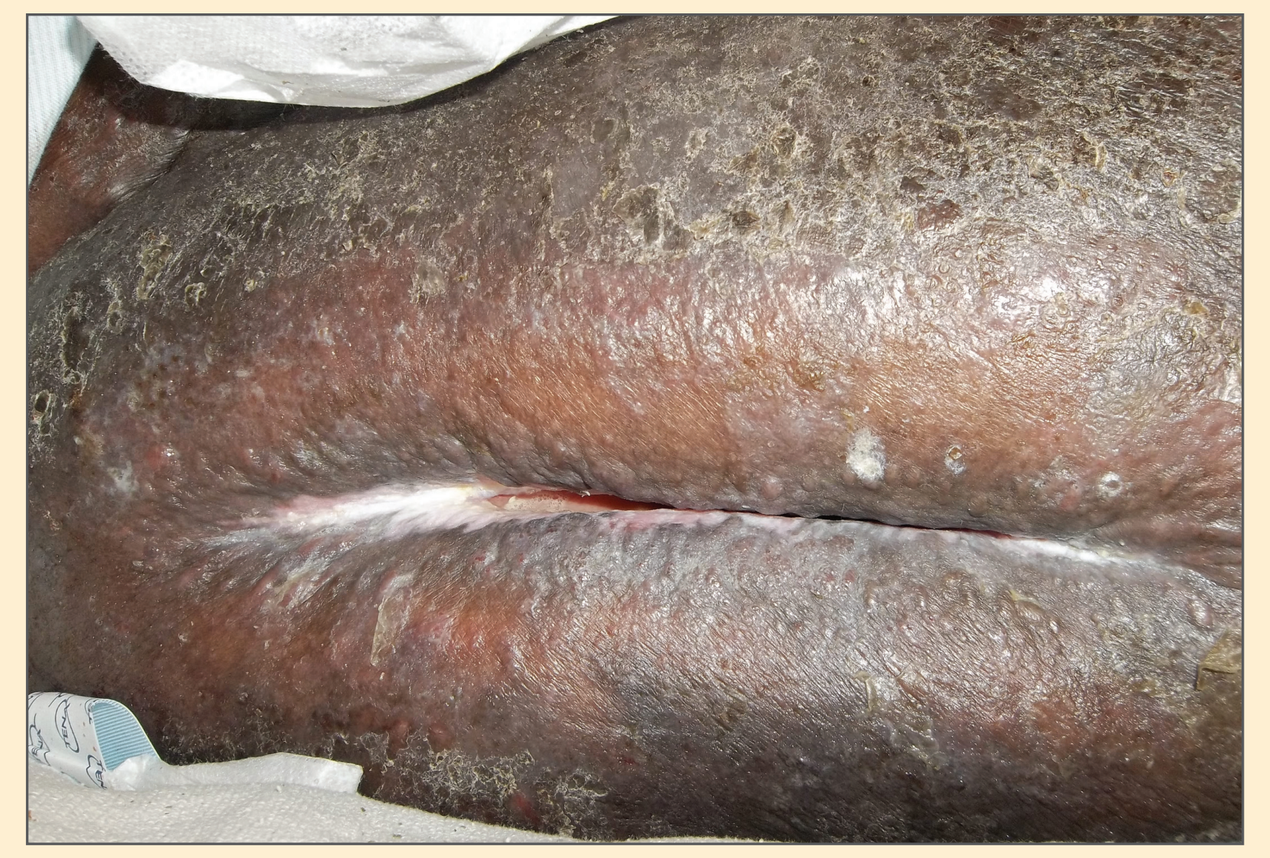
5/23/12

Date	Length	Width	Depth	Area (cm2)	Volume (cm3)	% Area Reduction	% Volume Reduction
2/8/12	13.7	0.7	3.2	7.53	24.10	Initial Measurement	Initial Measurement
2/15/12	14.0	1.0	3.4	11.00	37.39	-46.0%	-55.1%
2/22/12	12.7	1.7	3.2	16.96	54.26	-125.1%	-125.1%
2/29/12	13.0	1.4	3.1	14.29	44.31	-89.8%	-83.9%
3/6/12	13.0	1.0	2.6	10.21	26.55	-35.6%	-10.1%
3/12/12	11.7	0.7	2.2	6.43	14.15	14.6%	41.3%
3/19/12	11.0	1.2	2.4	10.37	24.88	-37.6%	-3.2%
3/28/12	11.0	1.0	2.1	8.64	18.14	-14.7%	24.7%
4/4/12	10.5	1.0	2.4	8.25	19.79	-9.5%	17.9%
4/11/12	9.8	0.8	2.2	6.16	13.55	18.2%	43.8%
5/23/12	8.5	0.4	1.6	2.67	4.27	64.5%	82.27%

CONCLUSION

ALH has shown to be an alternative to other costly wound treatment techniques. This data demonstrates situations of wounds not benefiting from NPWT but showing benefits within one week after the introduction of ALH. ALH is now considered at our facility earlier in the wound healing process rather than waiting until after other methods have failed or waiting until the wound has stalled.

References: References

1. Gethin G, Cowman S. Changes in surface pH when a honey dressing was used. Wounds UK Conference Proceedings; 13-15 November 2006. Wounds UK, Aberdeen Poster

2. Gethin G, Cowman S. Manuka honey vs. hydrogel – a prospective, open label, multicentre, randomised controlled trial to compare desloughing efficacy and healing outcomes in venous ulcers. 2008. *Journal of Clinical Nursing.*

*MEDIHONEY® Active Leptospermum Honey Dressing, Derma Sciences, Inc., Princeton, New Jersey.
*Xtrasorb® Super Absorbent Dressing, Derma Sciences, Inc., Princeton, New Jersey.

Poster content was updated from poster originally presented at SAWC Fall 2012, Baltimore, MD

0668721-1-EN

Derma Sciences provided an educational grant to support this research. The information may include a use that has not been approved or cleared by the Food and Drug Administration. This information is not being presented on behalf of Derma Sciences.