USE OF ACTIVE *LEPTOSPERMUM* HONEY TO PROMOTE AUTOLYTIC DEBRIDEMENT AND WOUND HEALING IN CHALLENGING CHRONIC WOUNDS

Margarita Simón, APRN, BC, CWCN, Simón Wound Consulting, PLLC / Kim Smith, Executive Director, Gentiva Home Health / Rachele Uptegrow, RN, Gentiva Home Health
Virginia Beach, VA

PURPOSE/INTRODUCTION

The care and management of patients with chronic wounds and their far-reaching effects challenge both the patient and the practitioner. In the current healthcare environment, clinicians are increasingly under pressure to use wound care products that can be used in a variety of wounds to achieve different outcomes, depending on the wound bed requirements.

METHODOLOGY

This series of 3 patients with 4 wounds focuses on topical wound care, in particular Active *Leptospermum* Honey* (ALH). The three patients involved were chosen due to the chronicity of the wounds despite other advanced methods of treatment including but not limited to bactroban, silicone foam dressings, cadexomer iodine gel**, doxycycline, and compression. The patients had multiple co-morbidities including dyslipidemia, obesity, mycosis fungoides, mastectomy, and venous insufficiency. The treatment plan was changed to ALH with an absorptive secondary dressing to assess if topical dressings like ALH will promote autolytic debridement and wound healing.

RESULTS

In evaluating the outcomes, it is evident that even with varied etiologies, wound age, and co-morbidities a common outcome was the reduction in necrotic tissue of at least 40% in two weeks with the addition of ALH, despite having been non-healing with previous topical dressing choices. Wound area also improved. All patients verbalized satisfaction with the improvement in the wounds.

CONCLUSION

Further research is encouraged to determine effect on additional wound types looking at rate of necrotic tissue removal and wound healing.

References: Bryant RA , Nix, D.P. Acute and Chronic Wounds: Current Management Concepts. St. Louis, MO: Mosby Elsevier; 2012. Pieper B. Honey-Based Dressings and Wound Care: An Option for Care in the United States. J of Wound, Ostomy and Continence Nursing 2009;36(1):60-66. (review)

*MEDIHONEY® Active *Leptospermum* Honey Dressings, Derma Sciences Inc., Princeton NJ
**IODOSORB gel ®, Smith and Nephew, London, UK
***MESALT®, Molnlycke, Gothenburg, Sweden
****AQUACEL Ag®, Systogenix, Gatwick, UK

Other cover dressing used during case series, included, but not limited to:
ALGISITE®, Smith and Nephew
AQUACEL® Ag++ rope, Conva Tec
OPTIFOAM®, Non-adhesive, Medline
ALLEVYN®, non-adhesive, Smith & Nephew
Maxorb® Extra CMC Alginate Dressings, Medline
Mepitel® Wound Contact Layer, Molnlycke Healthcare
Oil Emulsion Dressing Gauze, Non-Adherent, CURAD
TELFA® Non-Adherent Dressing, Covidien
Secura Protective Ointment, Smith & Nephew

Derma Sciences provided an educational grant to support this research. The information may include a use that has not been approved or cleared by the Food and Drug Administration. This information is not being presented on behalf of Derma Sciences.

Presented at SAWC Spring 2014, Orlando, FL

0668712-1-EN

CASE 1 – RIGHT LOWER EXTREMITY WOUND

Patient Outcomes: 14 days to closure

PMH: DVT and Venous Insufficiency

Case Discussion: 89 year old frail woman presented with chronic ulcers of the lower extremity for greater than 1 year in duration.

Past medical treatment included skin grafting to the area, oral antibiotics, cadexomer iodine dressings, and compression with elastic tube bandage. Patient's biopsy results revealed positive for *Acroangiodermatitis.*

On 10/8/13, ALH Paste was initiated, covered with a non-adherent dressing, foam dressing secured with gauze bandage roll and elastic tube bandage. Skin care for xerosis was stressed to prevent future skin dryness, cracking, and ulcers.

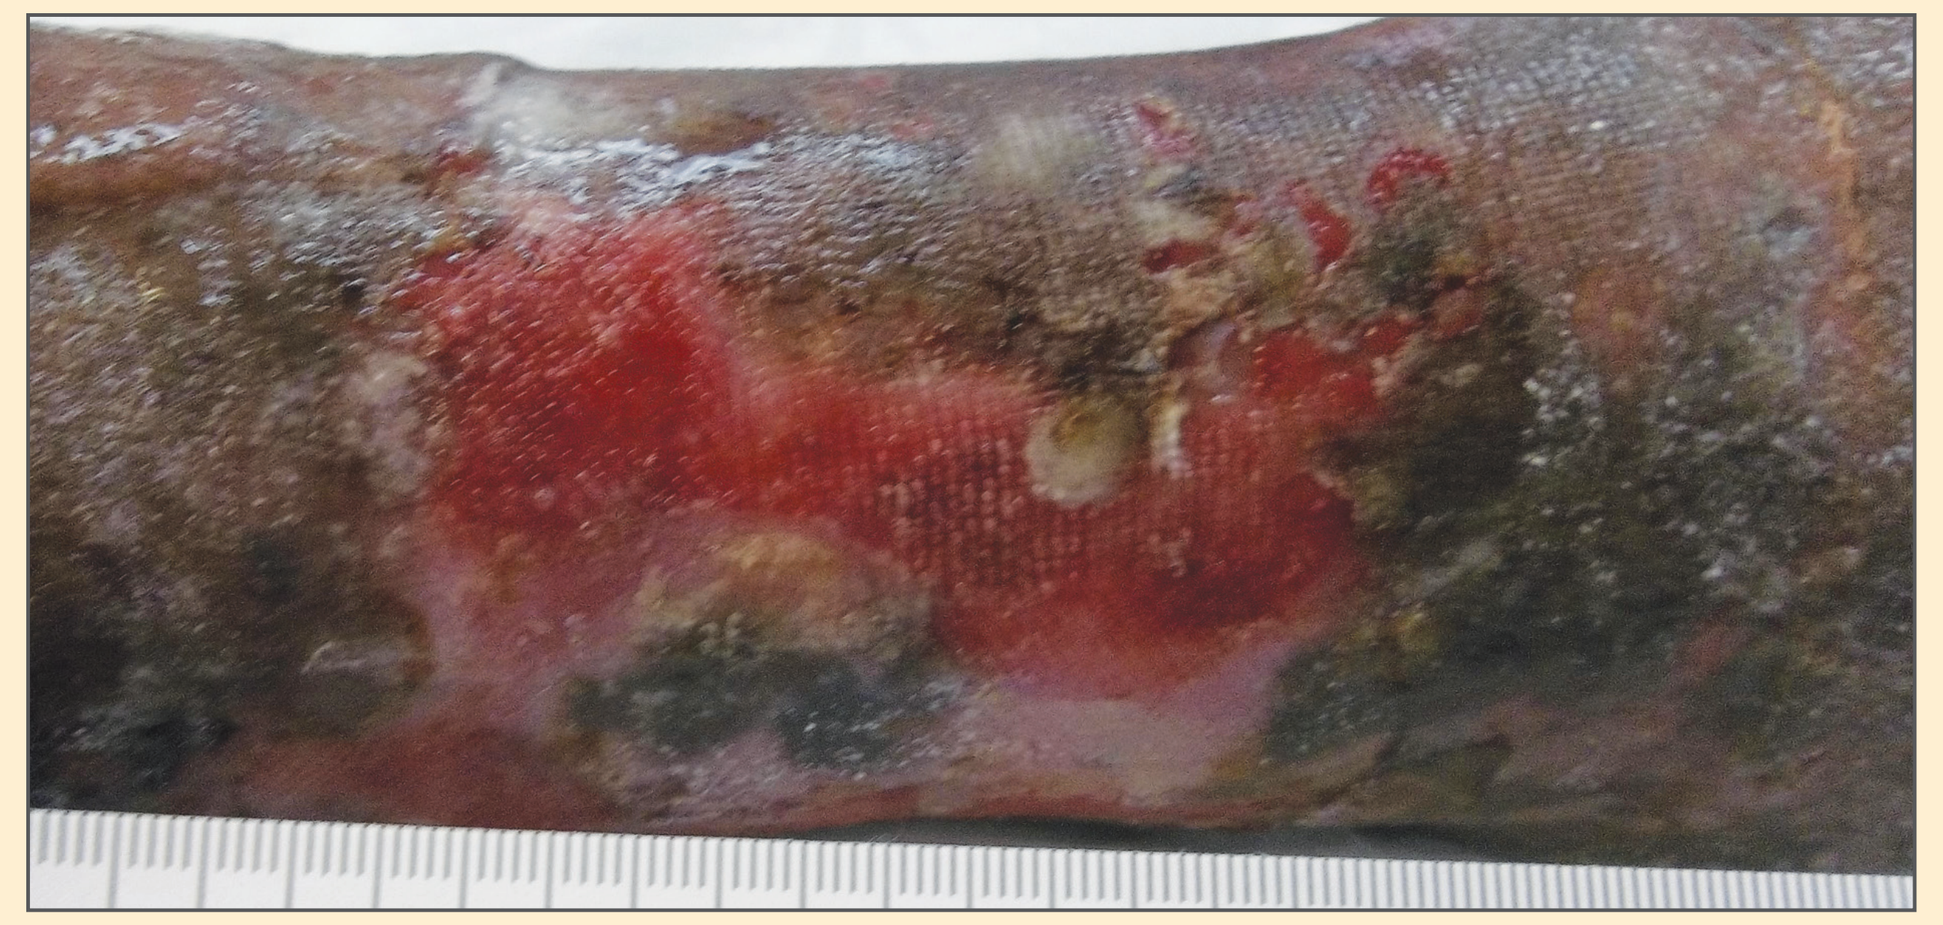

10/8/13 initial

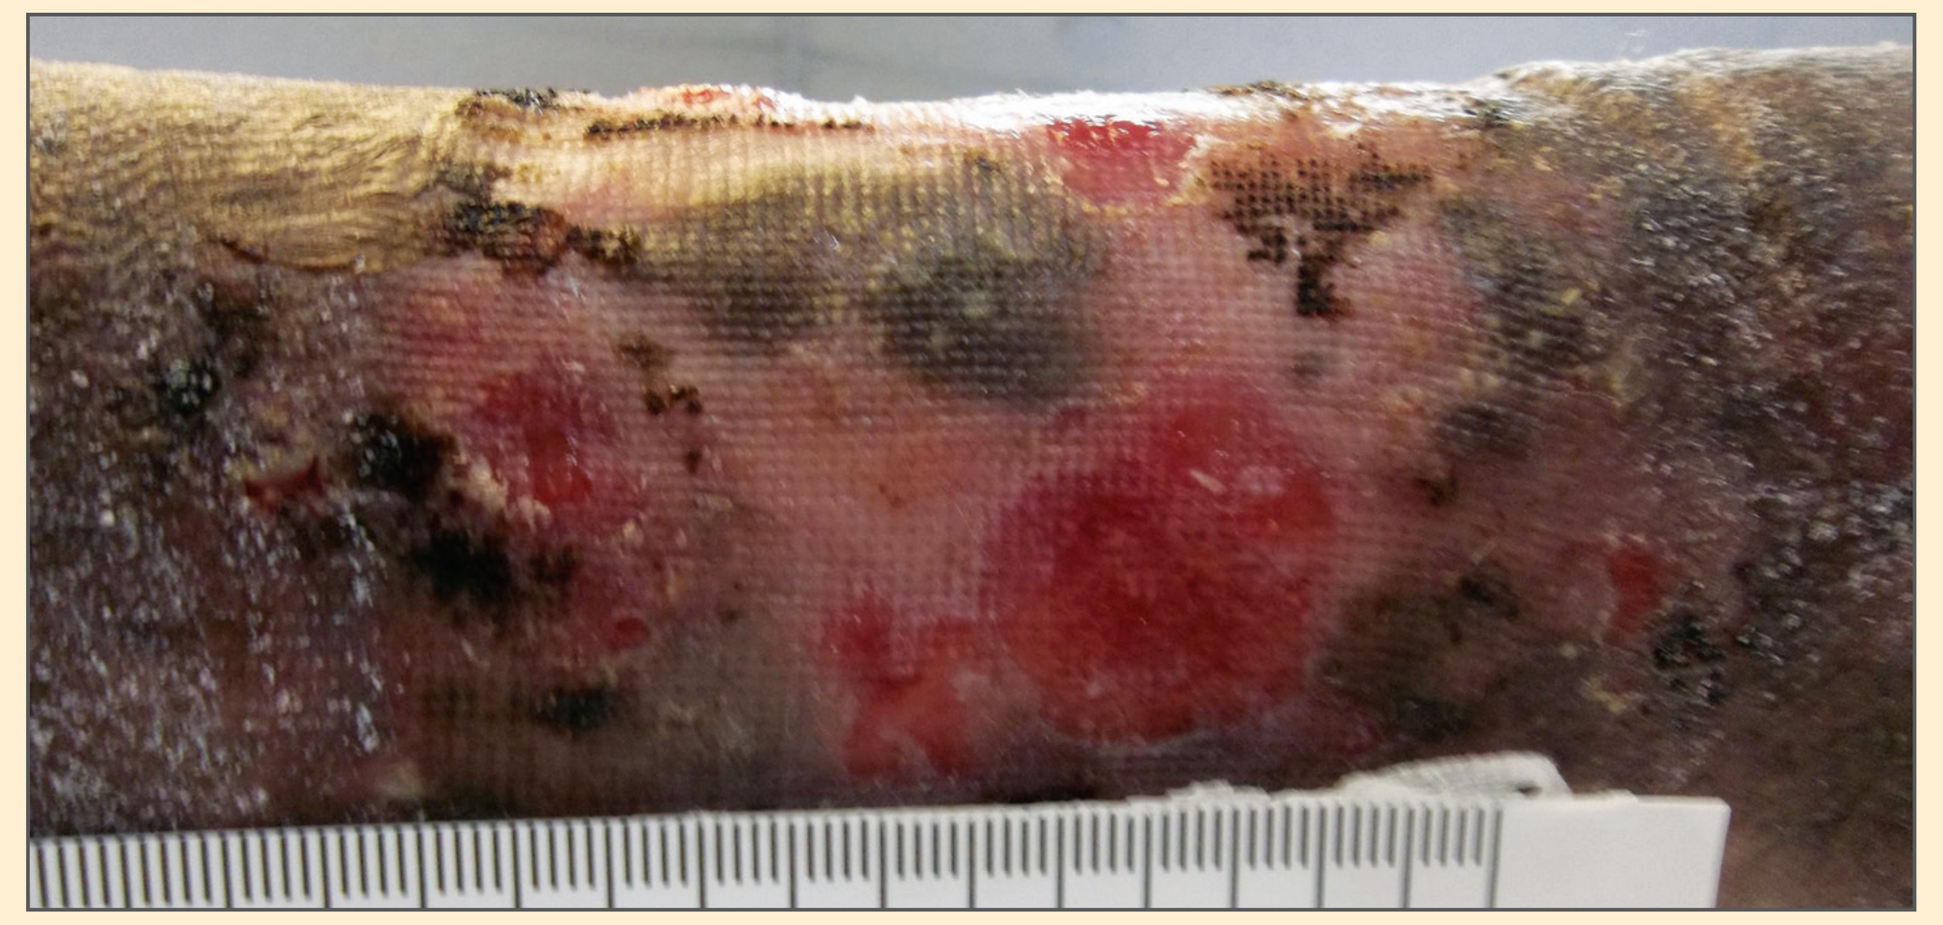

10/16/13

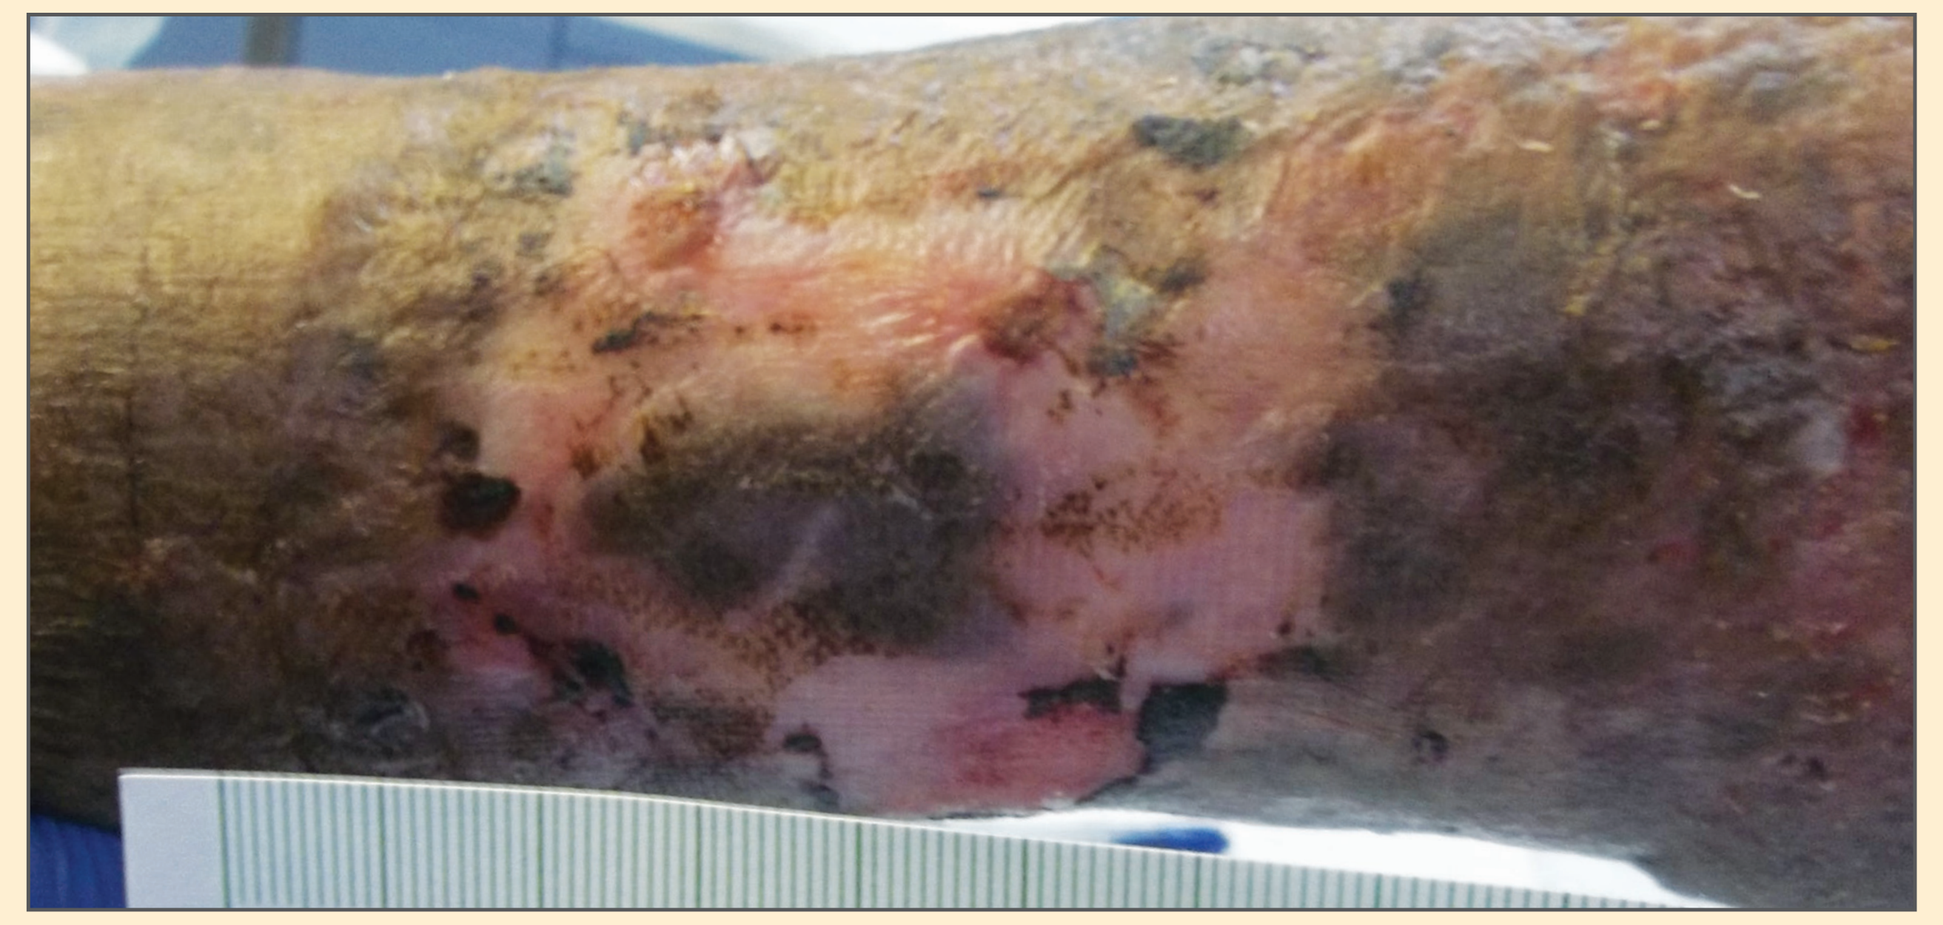

10/22/13 healed

CASE 2 – RIGHT MEDIAL/LATERAL ANKLE

Patient Outcomes: 70 days to closure for medial wound, 60 days to closure for lateral wound

PMH: HTN, DMII, Dyslipidemia, Obesity, Mycosis Fungoides since 2001. Wound present since a biopsy with large shave of the right medial ankle in the fall of 2012. History of MRSA treated with co-trimoxazole DS 2 BID for 3 months.

Case Discussion: 45 year old presented with wounds to right ankle – medial and lateral on 7/23/13. Upon evaluation of admission to homecare, patient's wound was found to be significantly painful even with slight touch and temperature intolerance. Wound bed mostly necrotic with thick patches of dry skin at peri-wound/leg/foot.

7/23/13 Wound Treatment changed to: Cleanse with NS (warmed), apply ALH Paste on Calcium Alginate, cover with non-adherent gauze, skin barrier cream to peri-wound skin. Apply a skin barrier cream to the peri-wound/leg/feet, telfa, and secured with roll gauze. Due to the large amount of drainage, daily dressings were done through 7/30/13. As drainage decreased on 7/30/13 to 8/17/13 dressings were changed every other day, then to 3x a week.

Patient made progress in several areas: Less pain with wound care, peri-wound dryess resolving, necrotic tissue significantly being debrided, no infection and patient has not had the need for antibiotics since the start of care. Blood sugars are also under better control.

MEDIAL WOUND

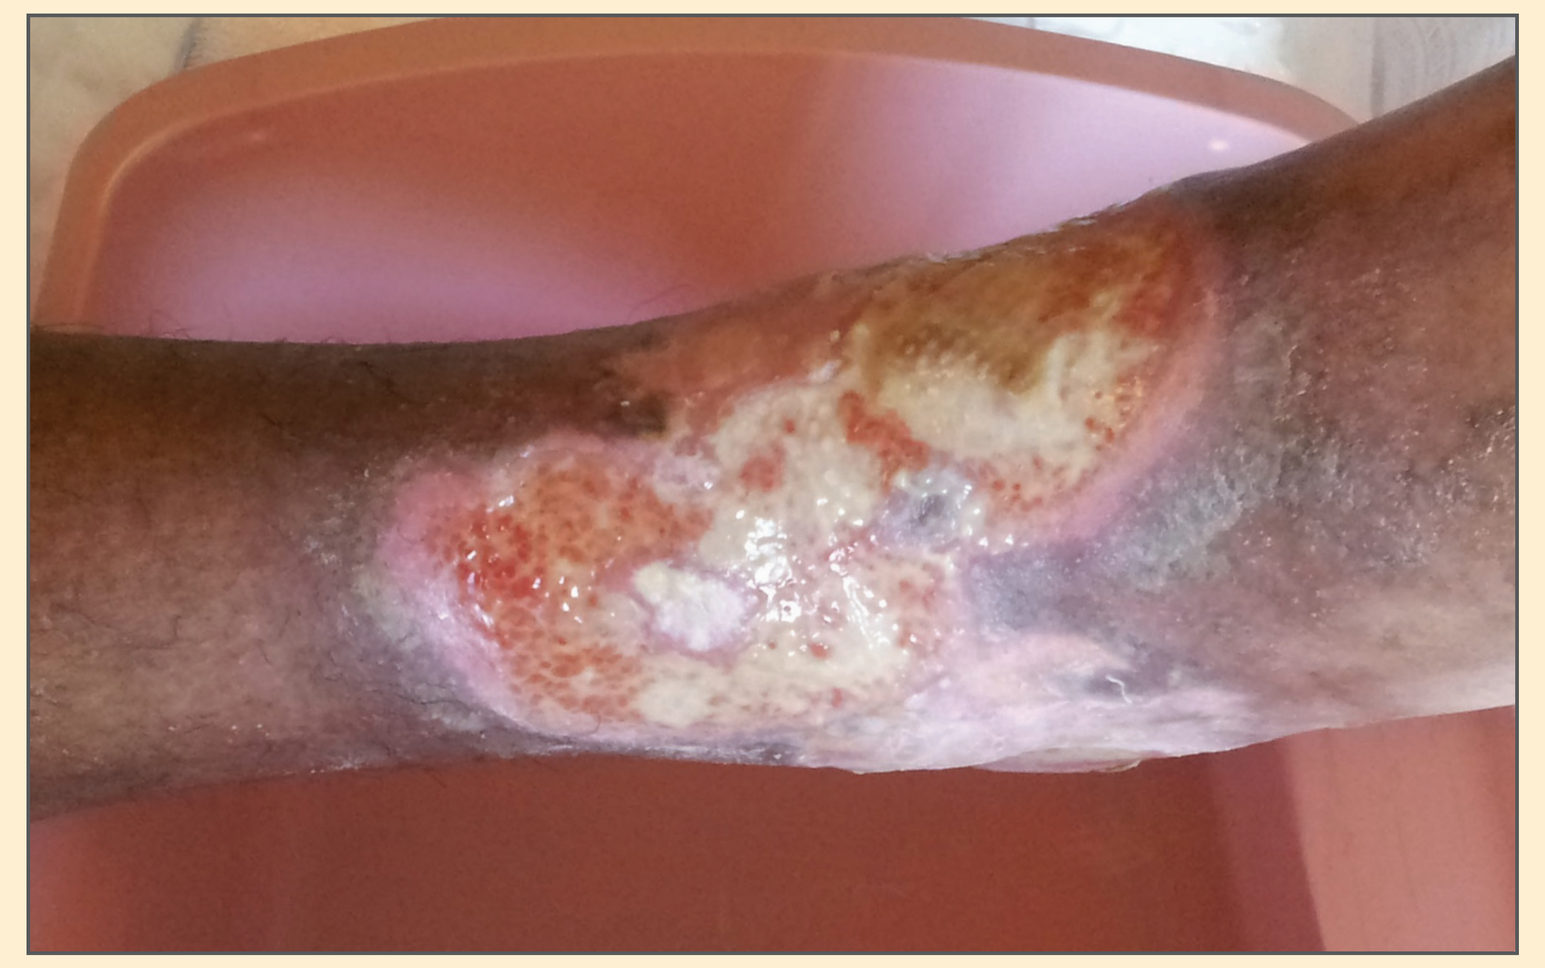

7/23/13

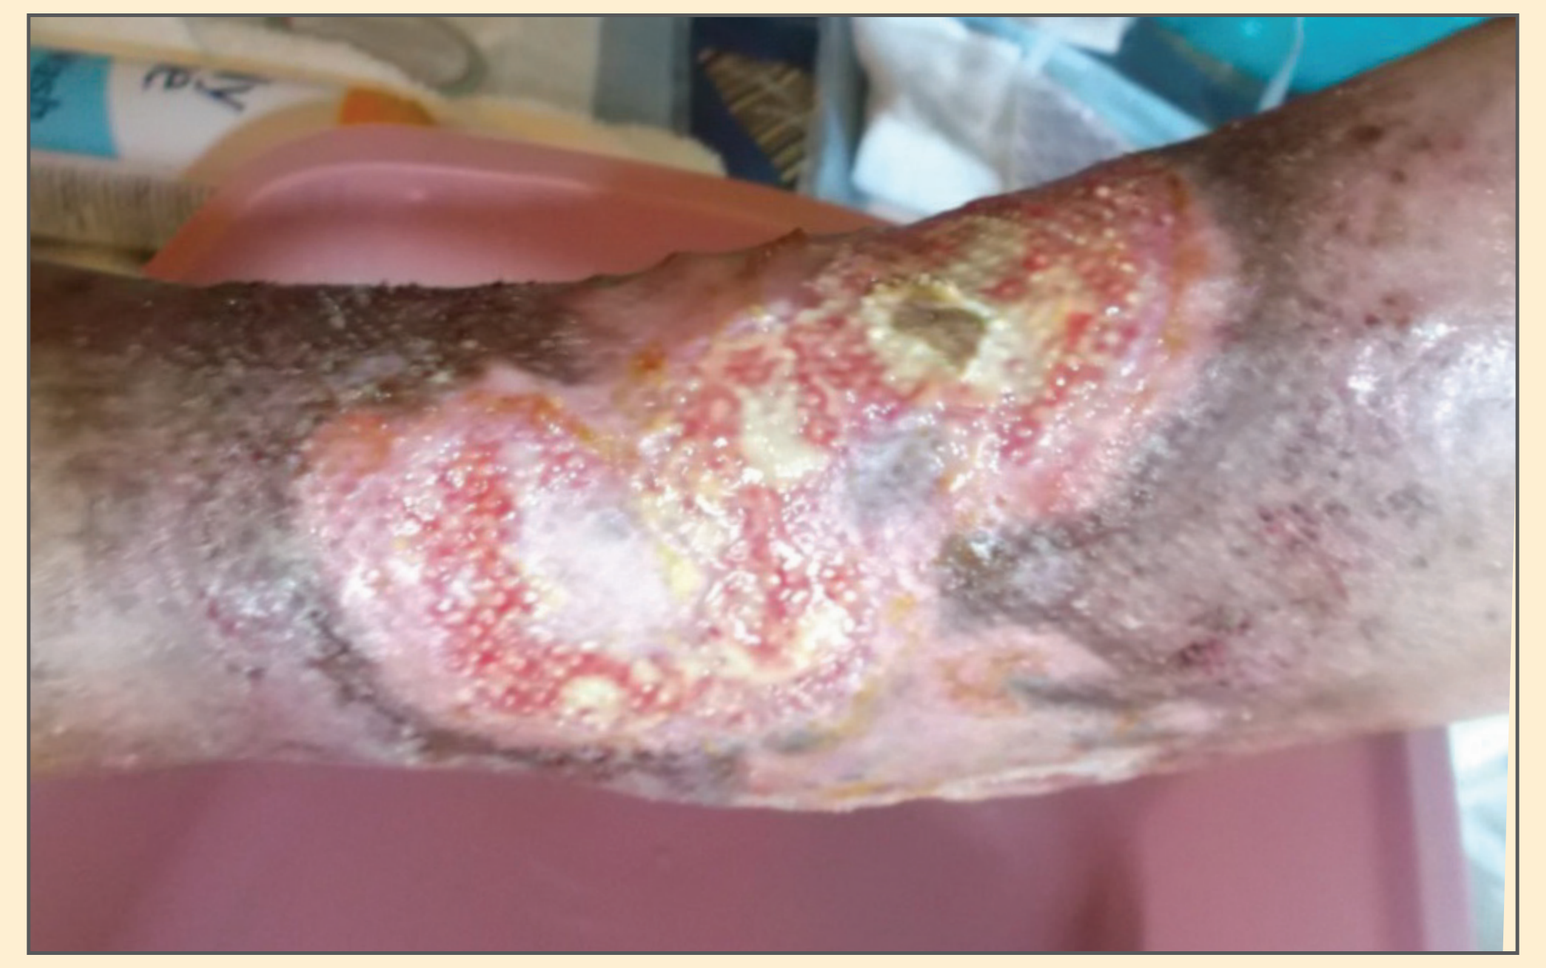

7/30/13

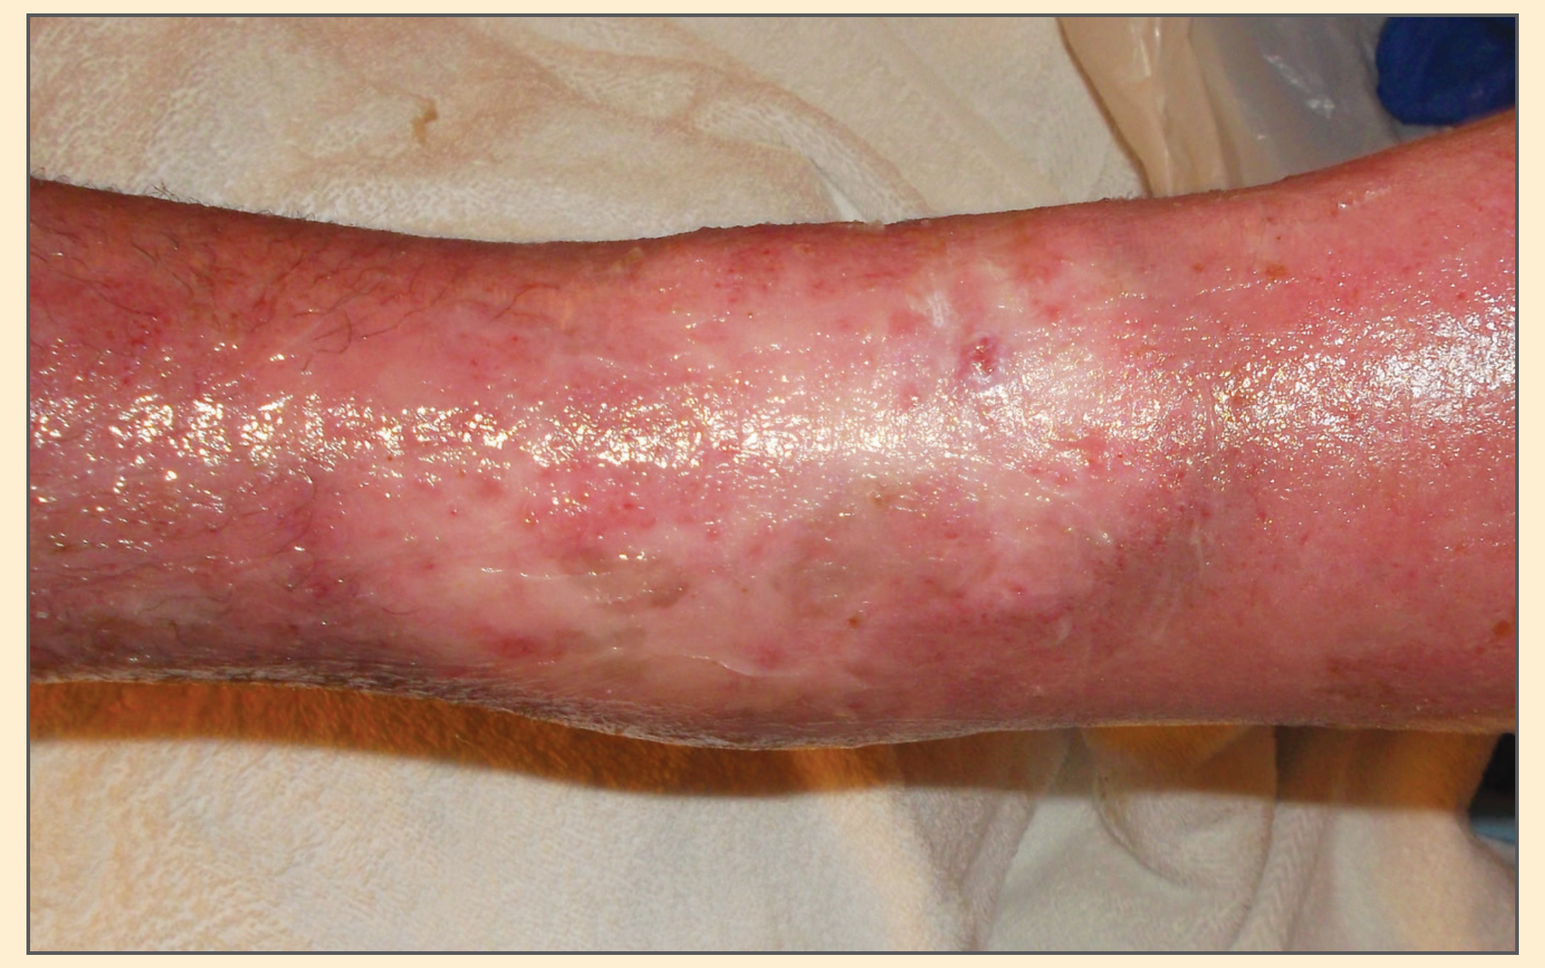

10/01/13

LATERAL WOUND

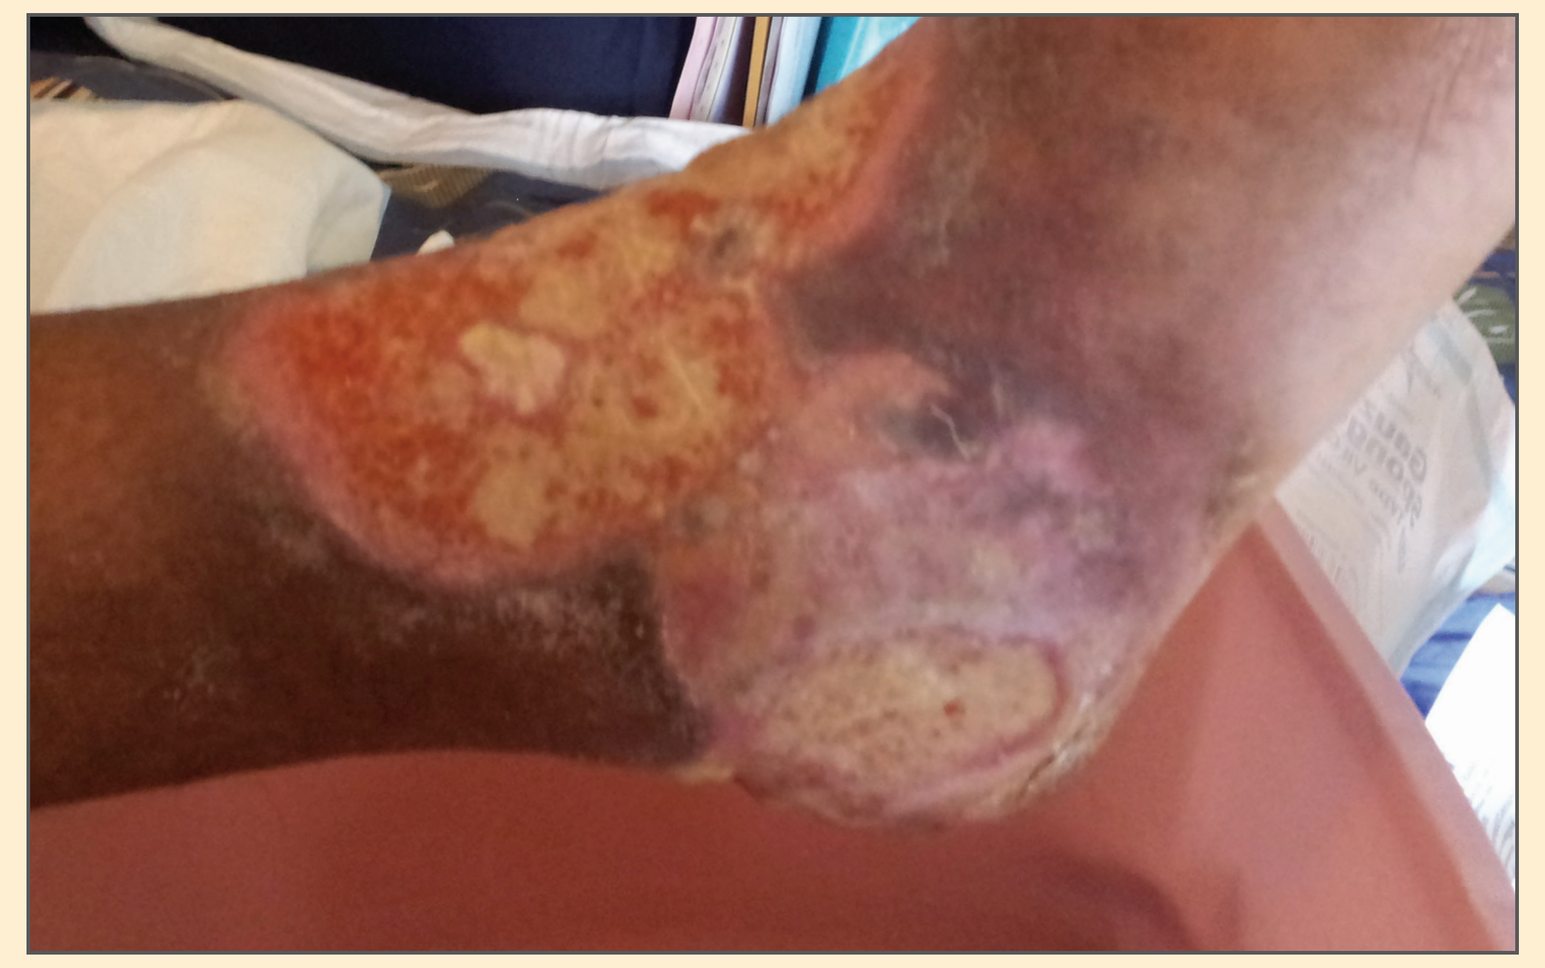

7/23/13

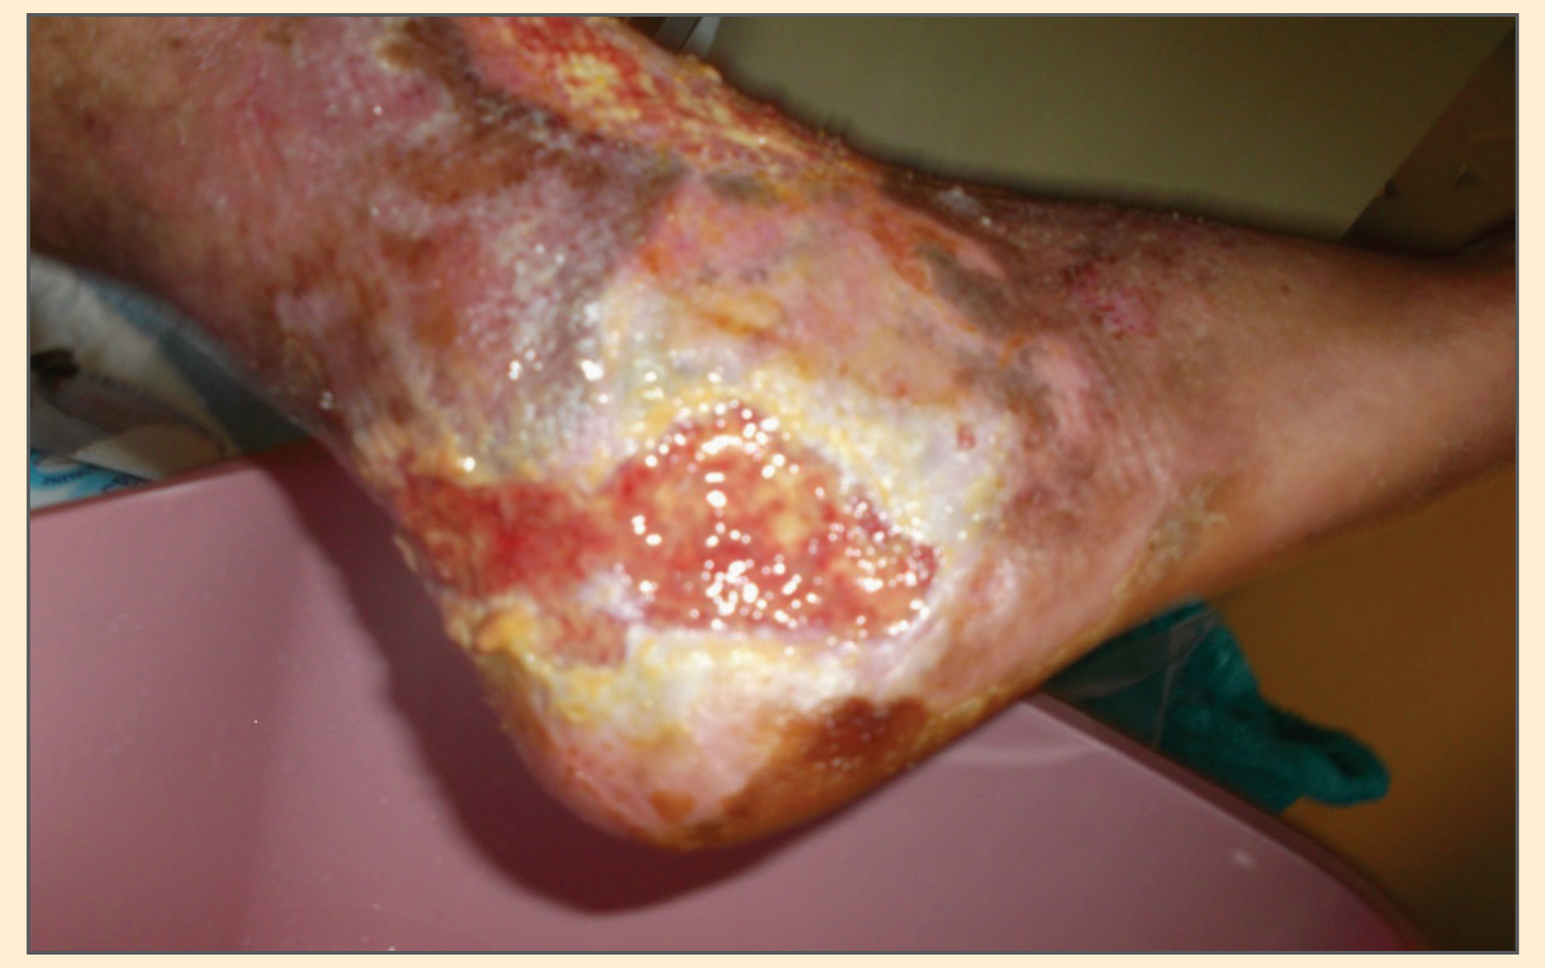

7/30/13

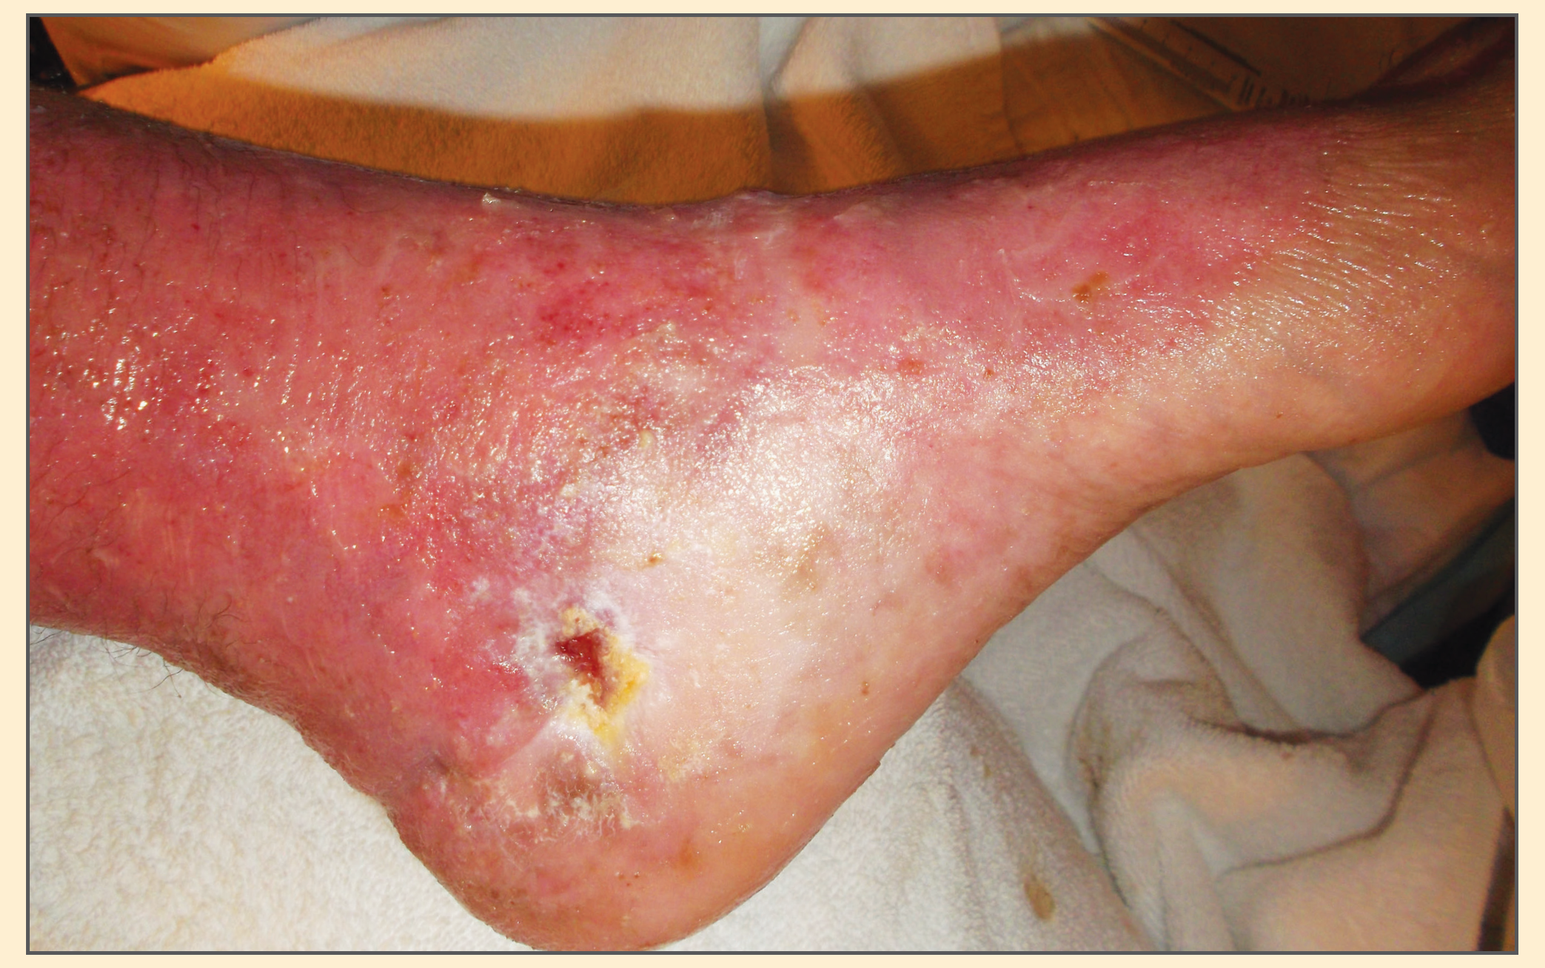

10/01/13

CASE 3 – ABDOMINAL WOUND POST TRAM FLAP RECONSTRUCTION

Patient Outcomes: 49 days to re-epitheliazation

PMH: Mastectomy 1997 with TRAM flap reconstruction, atrial fibrillation with cardioversion, and depression.

Case Discussion: 5/1/13 patient was referred to the Wound Care Center with abdominal wound present for some time before she saw her PCP who referred her to the Wound Care Center. Patient presented with abdominal wall cellulitis and pain. The wound bed surrounding the umbilicus was covered with eschar and malodorous drainage. The eschar was crosshatched and started on ALH Gel, covered with sodium impregnated gauze*** moistened with saline. Silver impregnated calcium alginate was packed into the umbilicus****. Barrier ointment was applied to the peri-wound, area was covered with ABD, and secured with tape.

Complete resolution of the abdominal wall wound was achieved on 6/19/13. ALH was used until closure.

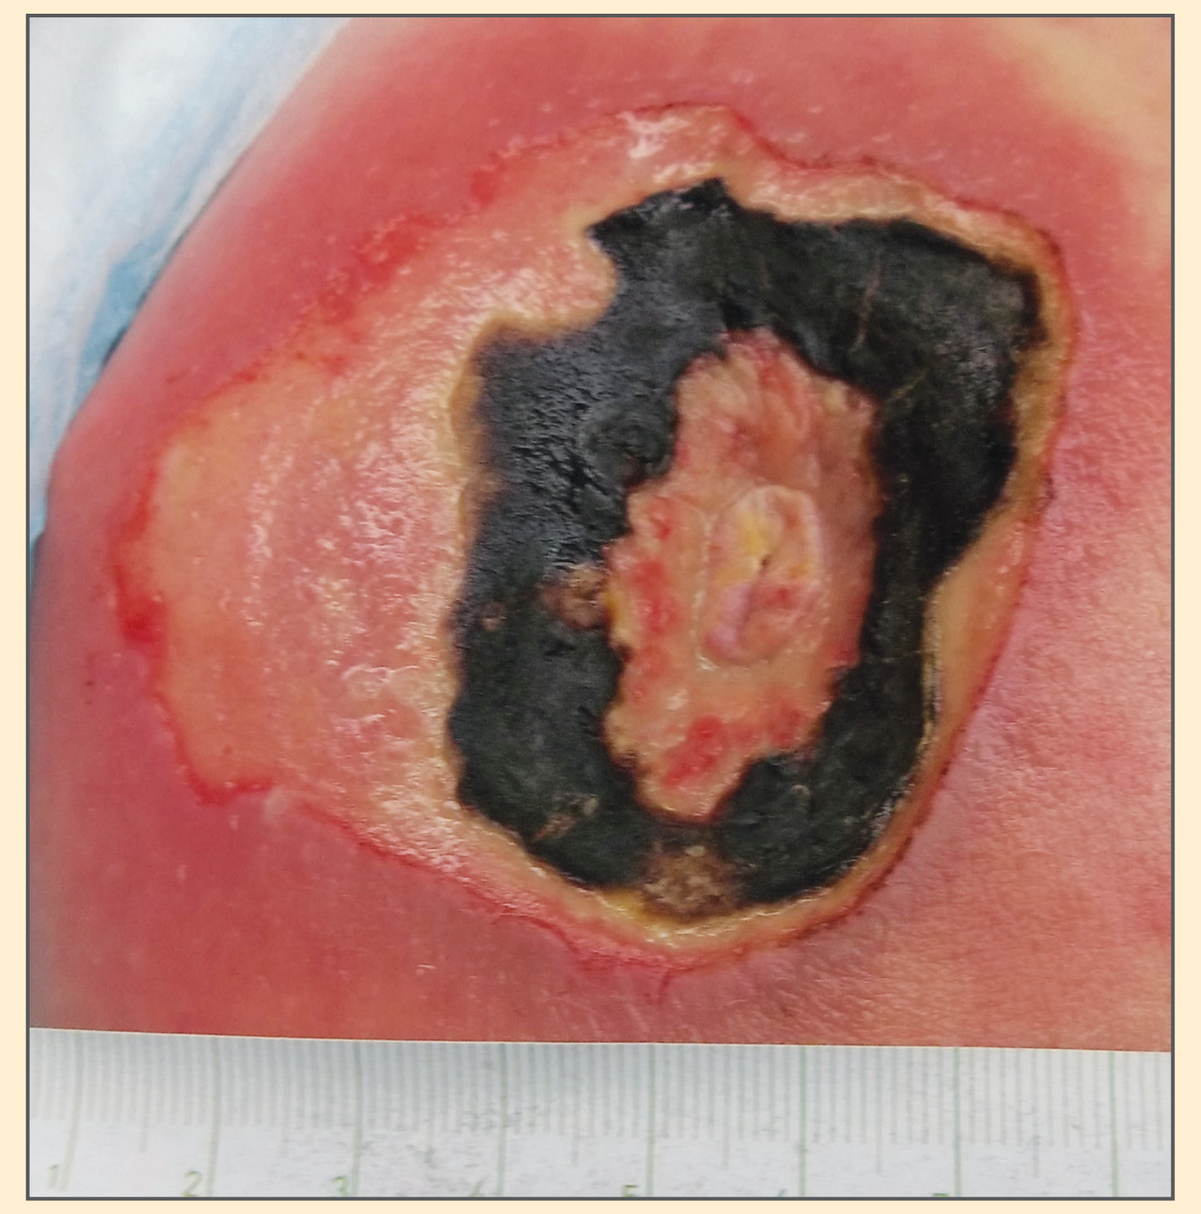

5/1/13 initial photo

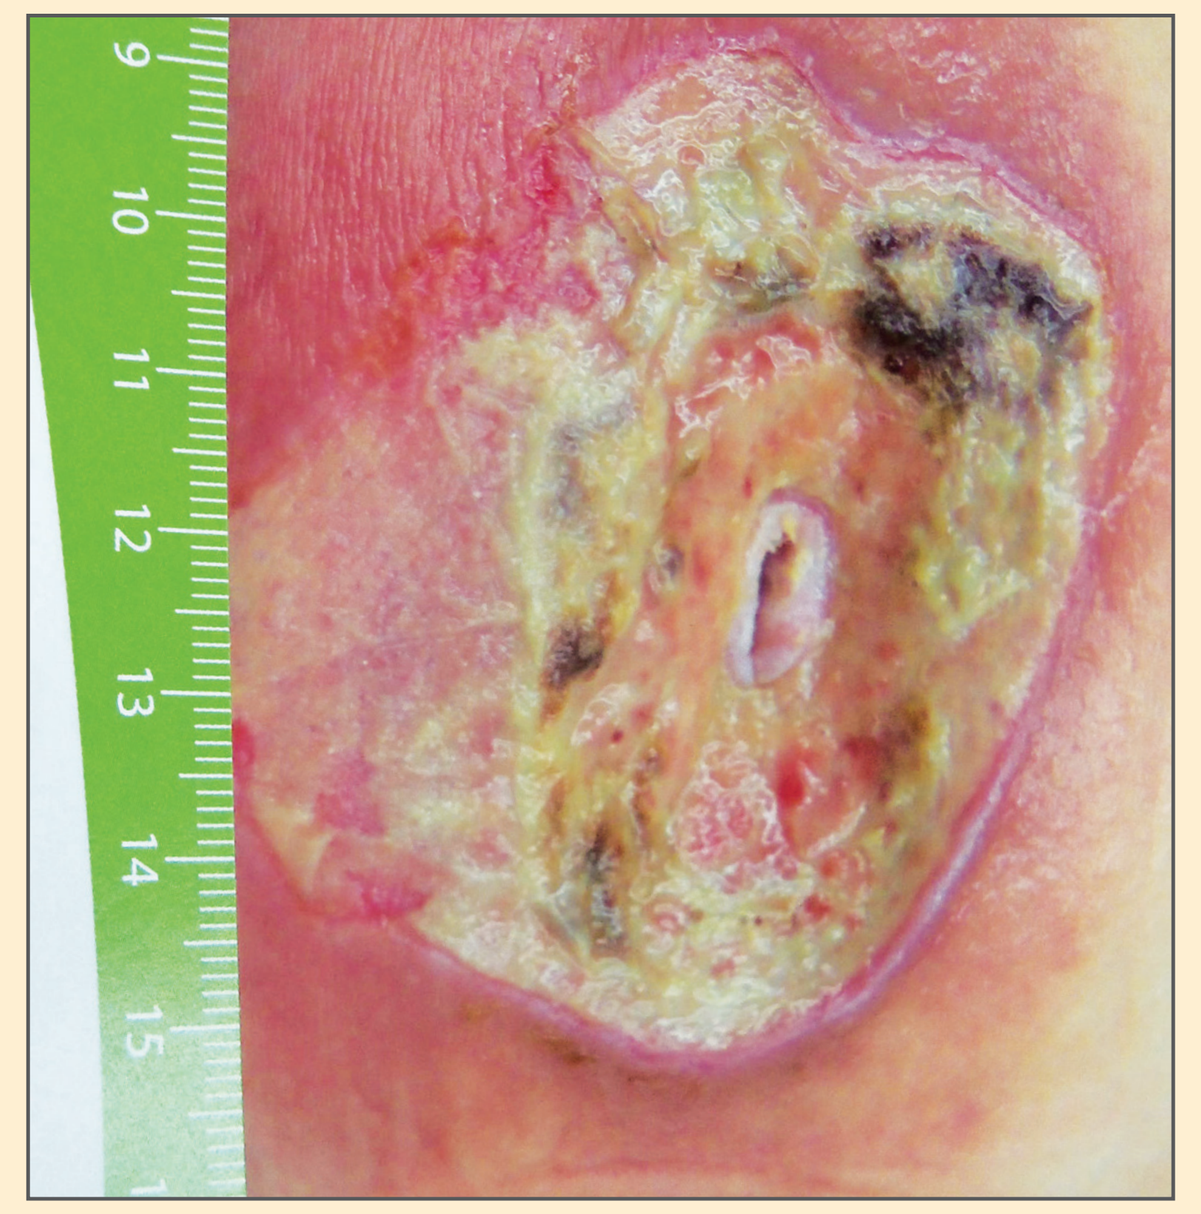
9 10 11 12 13 14 15

5/3/13

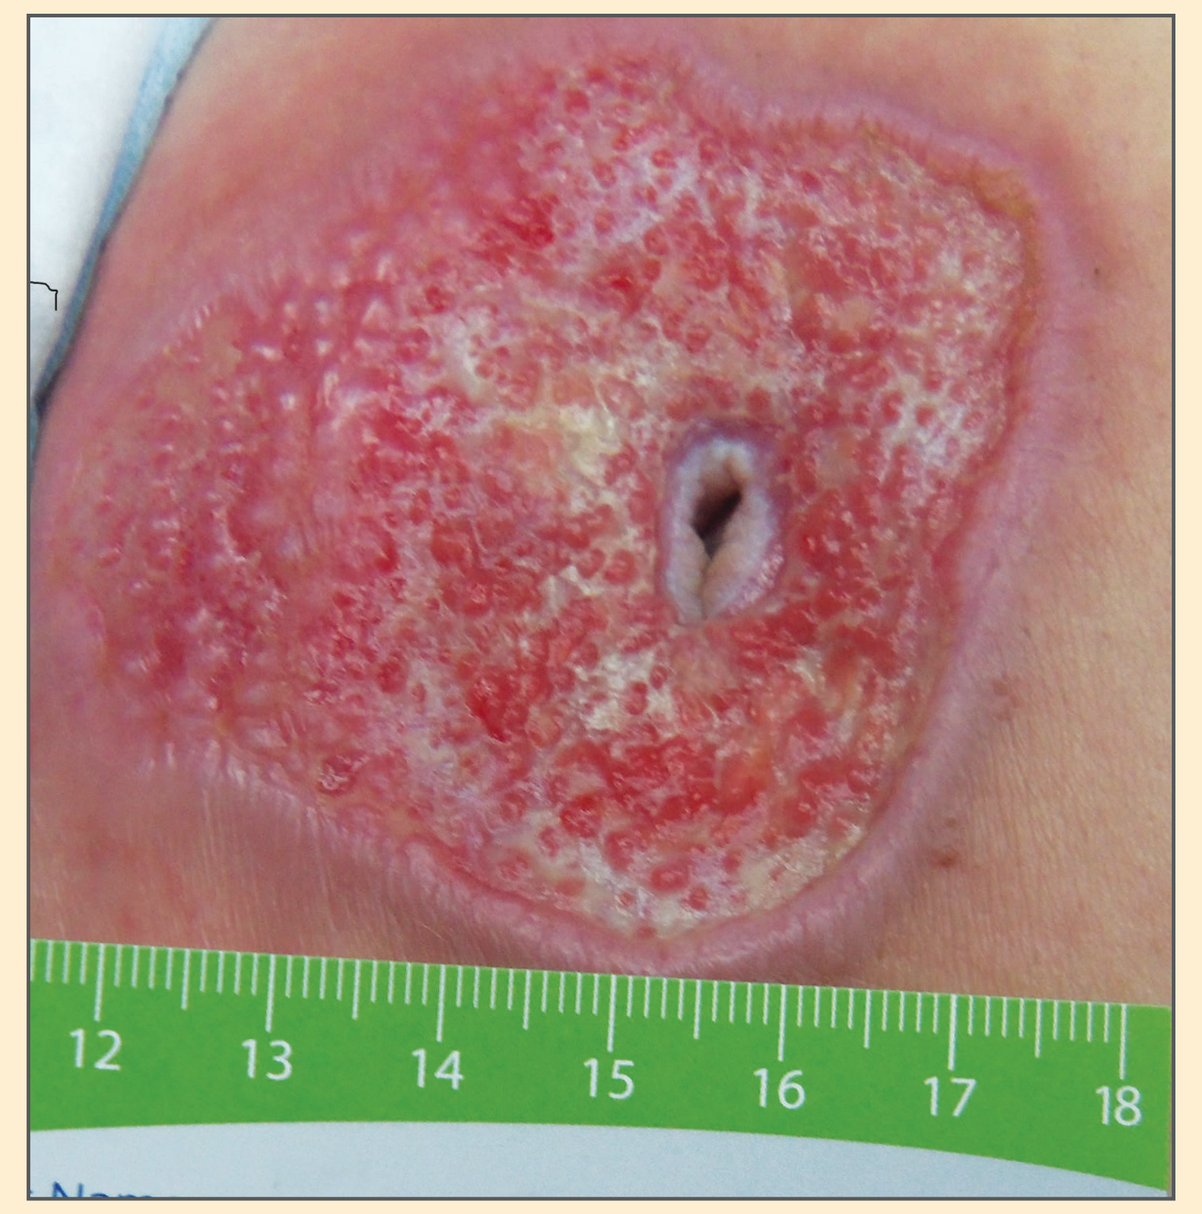
12 13 14 15 16 17 18

5/10/13

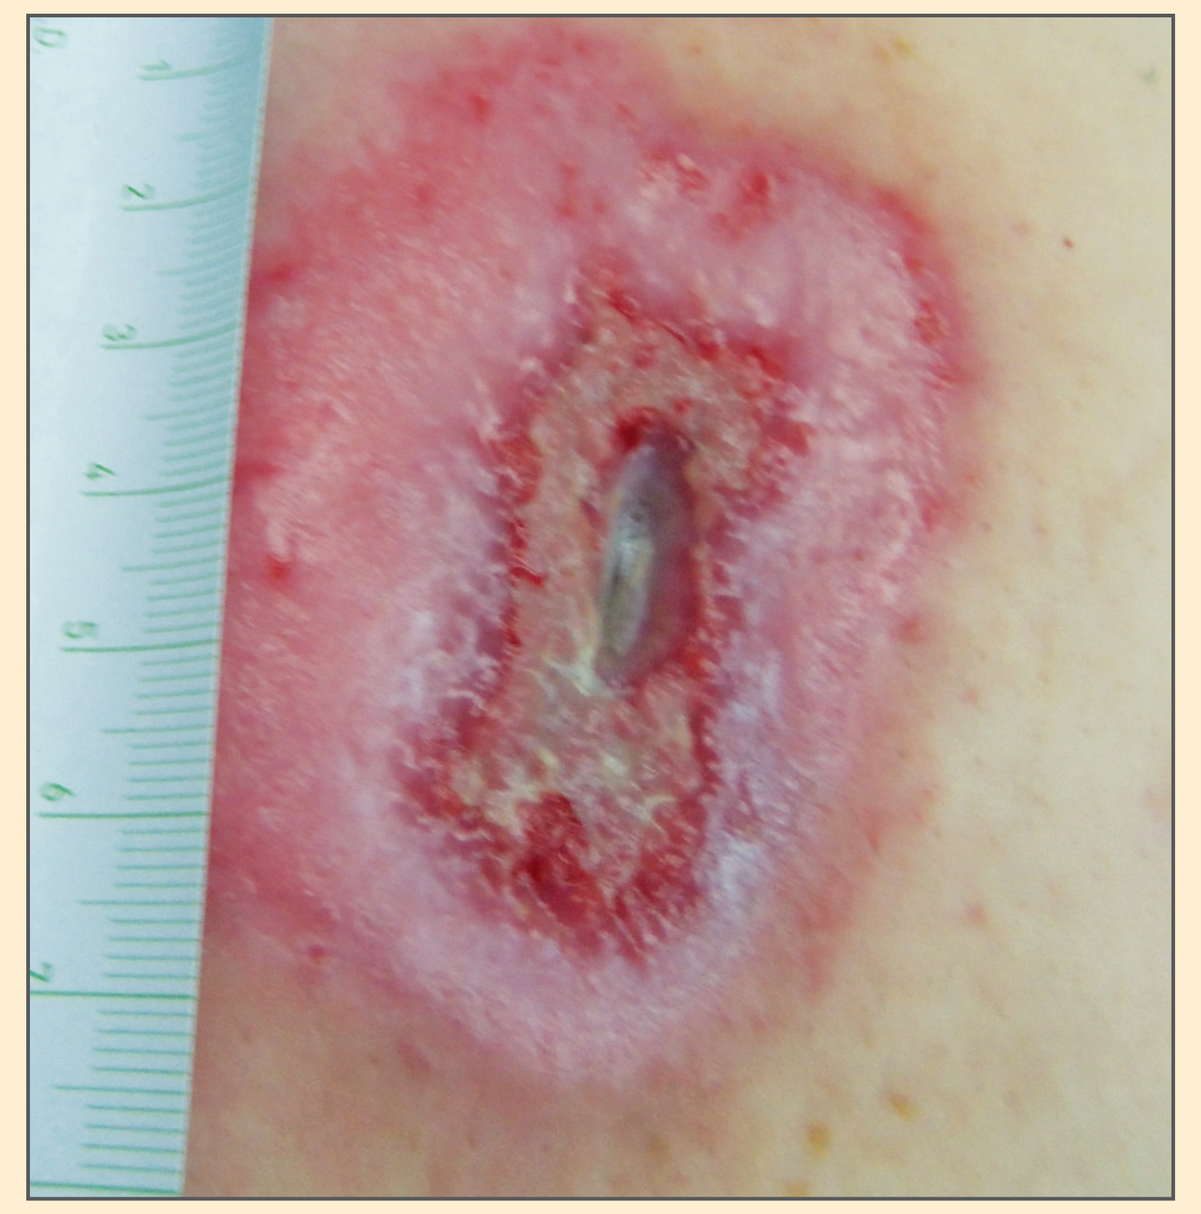

5/29/13

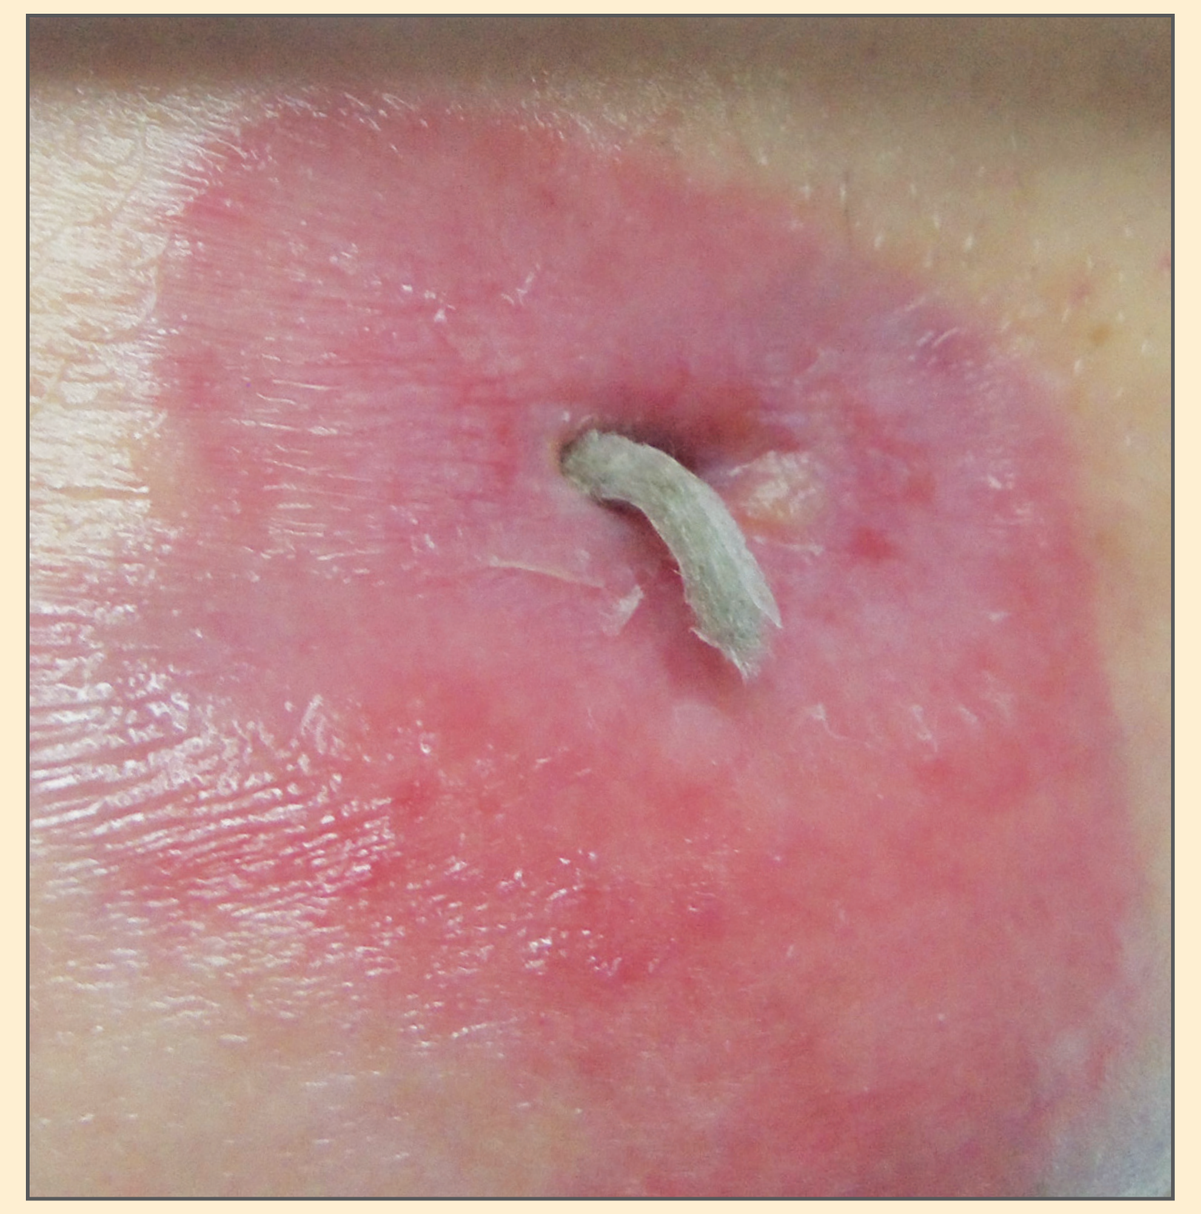

6/19/13